Interim follow-up analysis in the REP 401 protocol: Functional control of HBeAg negative chronic HBV infection persists after withdrawal of combined therapy with REP 2139 or REP 2165, tenofovir disoproxil fumarate and pegylated interferon α-2a

M. Bazinet[1], V. Pântea[2], G. Placinta[2], I. Moscalu[3], V. Cebotarescu[2], L. Cojuhari[2], P. Jimbei[4], L. Iarovoi[2], V. Smesnoi[4], T. Musteata[4], A. Jucov[2,3], A. Krawczyk[5] A. Vaillant[1]

1. Replicor Inc. Montreal, Canada
2. Department of Infectious Diseases, Nicolae Testemitanu State University of Medicine and Pharmacy, Chișinău, Republic of Moldova
3. ARENSIA Exploratory Medicine, Republican Clinical Hospital, Chișinău, Republic of Moldova
4. Toma Ciorbă Infectious Clinical Hospital, Chișinău, Republic of Moldova.
5. Universitätsklinikum Essen, Institute for Virology, Essen, Germany.

AASLD THE LIVER MEETING® 2017 WASHINGTON, DC OCTOBER 20-24

Infectious Disease Clinic Toma Ciorbă Hospital

Universitätsklinikum Essen

INTRODUCTION

HBV subviral particles (SVPs) constitute more than 99.99% of circulating HBsAg. SVPs are derived from cccDNA and from integrated HBV DNA and can thus be produced in cccDNA-independent fashion. The nucleic acid polymer (NAP) REP 2139 blocks the assembly and release of HBV subviral particles (SVPs) derived from cccDNA and integrated HBV DNA, allowing the efficient clearance of serum HBsAg. This clearance facilitates the restoration of functional control of HBV infection by immunotherapy in HBV and HBV / HDV-co-infection[1,2]. The REP 401 protocol (NCT02565719) is a randomized, controlled trial assessing the safety and efficacy of REP 2139 and a REP 2139 derivative with improved tissue clearance (REP 2165) in combination with tenofovir disoproxil fumarate (TDF) and pegylated interferon alpha-2a (peg-IFN) in patients with chronic HBeAg negative HBV infection.

MATERIAL & METHODS

Twenty four weeks of lead-in TDF (300mg PO qD) was followed by randomization (1:1) into experimental and control groups (Table 1). The experimental group received 48 weeks of TDF (300mg PO qD), peg-IFN (180ug SC qW) and REP 2139-Mg or REP 2165-Mg (1:1, 250 mg IV infusion qW) (Table 2). The control group was to receive 48 weeks of TDF + peg-IFN but all patients have crossed over to 48 weeks of experimental therapy in the absence of a 3 log drop in HBsAg after 24 weeks of peg-IFN (see figure below). Viremia is monitored on the Abbott Architect and Realtime platforms.

Pre-treatment demographics

Parameter		Adaptive comparator control (TDF + peg-IFN)	Experimental (TDF + peg-IFN + NAPs)
Age (average / median)		36.9 / 36	38.6 / 39.5
Sex		27M / 3F	26M / 4F
HBV genotype	A	1	2
	D	19	18
Metavir score (based on Fibroscan)	F0-F1	12	10
	F2	3	6
	F2-F3	0	3
	F3-F4	3	1
Virologic baseline (average / median)	HBV DNA (IU/mL)	$3.6x10^7$ / $8.7x10^4$	$4.8x10^6$ / $4.8x10^4$
	HBsAg (IU/mL	14775.7 / 9302.5	9018.1 / 8743
	Anti-HBs (mIU/mL)	0.78 / 0.1	2.778 / 0.1
ALT (U/L, average / median)		71.65 / 49	91.95 / 56.5

REP 2139 versus REP 2165

NAP	Sequence 5' - 3'	Stability in human plasma 7 days @ 37°C (% of standard)	
		Neutral	Acidified
REP 2139	ACACACACACACACACACACACACACACACACACACACAC	93	86
REP 2165	ACACACACACACACACACACACACACACACACACACACAC	89	36

A = 2' O-methyl ribose modification in REP 2139 is 2'OH in REP 2165, allowing greater susceptibility to endonuclease attack and degradation.

REP 401 design

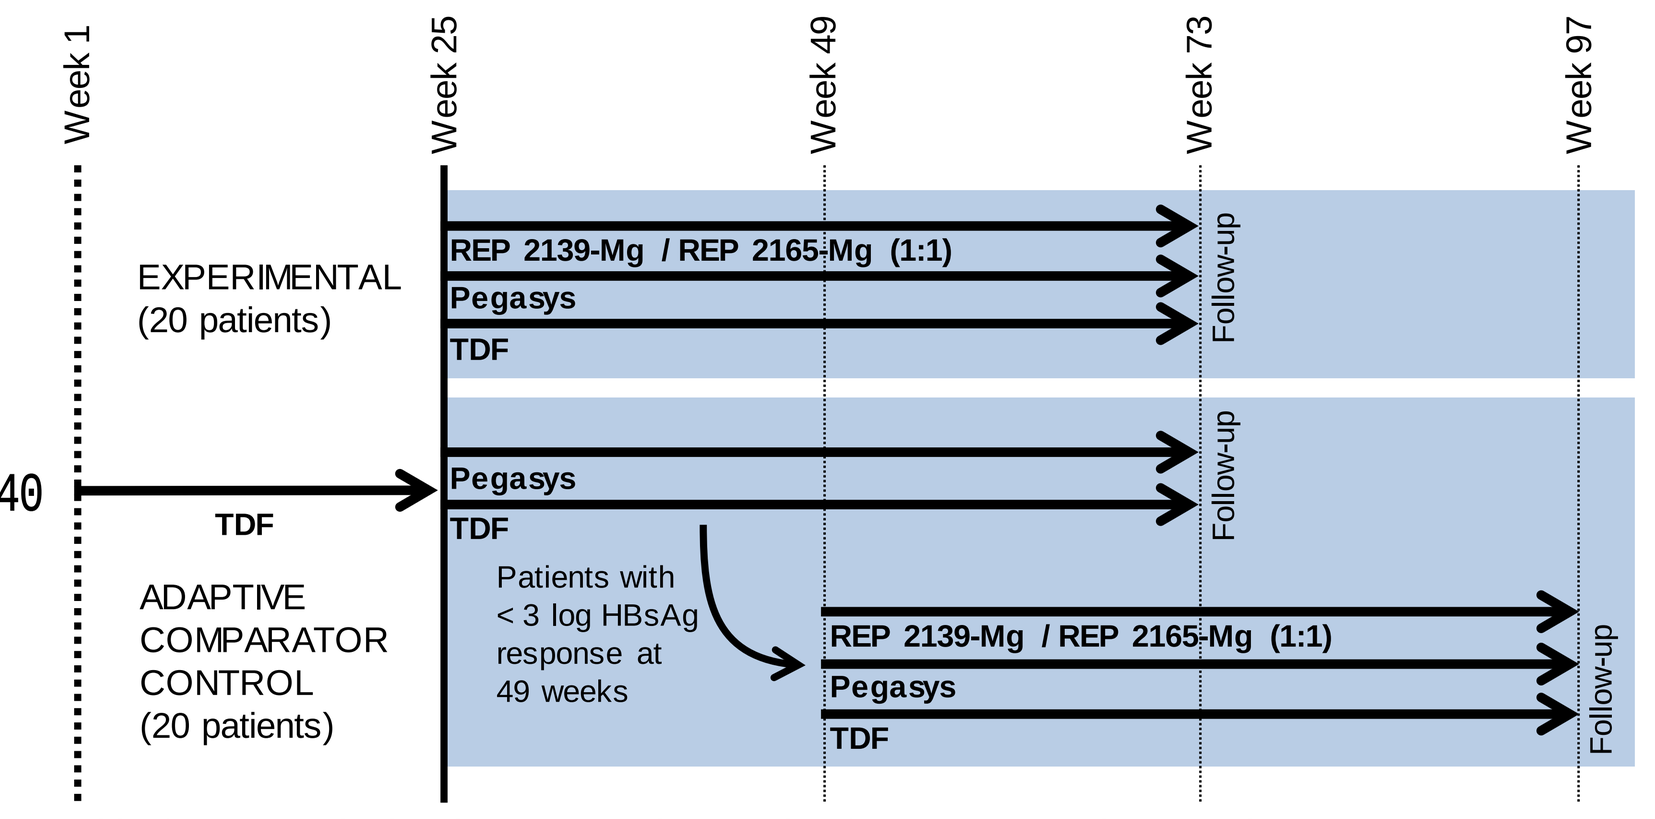
Week 1
Week 25
Week 49
Week 73
Week 97
EXPERIMENTAL
(20 patients)
REP 2139-Mg / REP 2165-Mg (1:1)
Pegasys
TDF
Follow-up
40
TDF
ADAPTIVE
COMPARATOR
CONTROL
(20 patients)
Patients with
< 3 log HBsAg
response at
49 weeks

Initial follow up scheduled 4, 12, 24 and 48 weeks after all treatment is stopped

DISCLOSURES

MB and AV are employees and shareholders in Replicor Inc. All other authors have no conflicts to declare.

INTERIM RESULTS (TREATMENT)

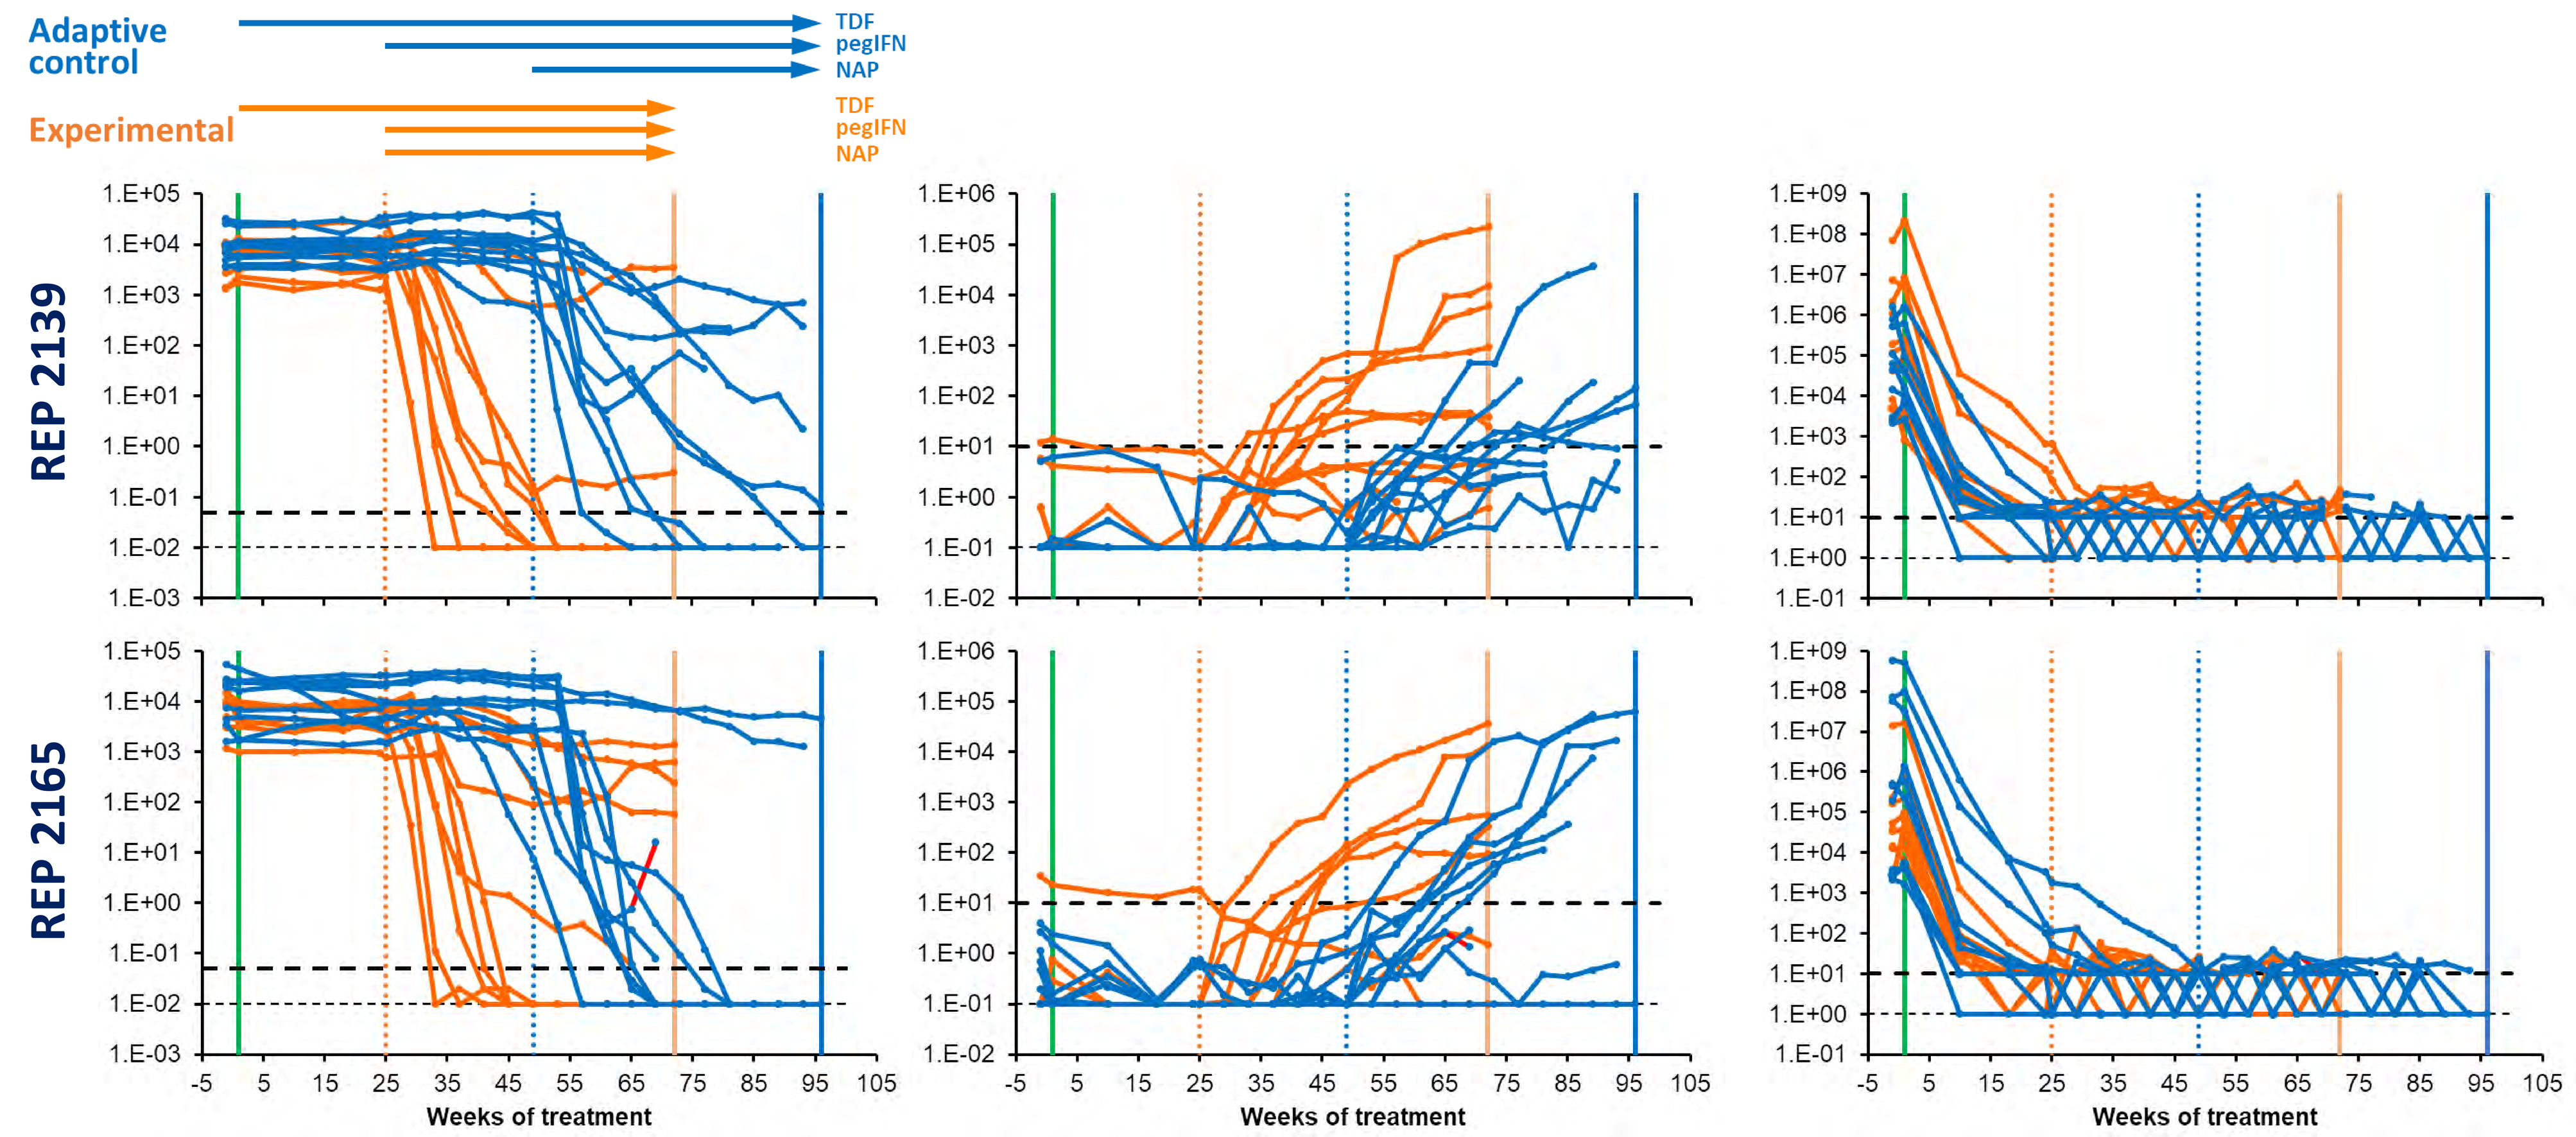
Adaptive control
Experimental
TDF
pegIFN
NAP
REP 2139
REP 2165
Weeks of treatment

Serum HBsAg (IU/mL) Serum anti-HBs (mIU/mL) Serum HBVDNA (IU/mL)

Viral dynamics

Individual analyses of HBsAg, anti-HBs and HBV DNA during treatment in the adaptive control (blue) and experimental (orange) groups are provided for patients receiving REP 2139 (top) and REP 2165 (bottom). Bold dashed horizontal lines indicate LLOQ (HBsAg, HBV DNA) or 10mIU/mL (anti-HBs). Fine dashed horizontal lines indicate target not detected (HBsAg and HBV DNA) or no significant anti-HBs present (< 0.1 mIU/mL). Red segment indicates off-treatment rebound in a single control patient who withdrew from the trial for depression attributed to Pegasys exposure.

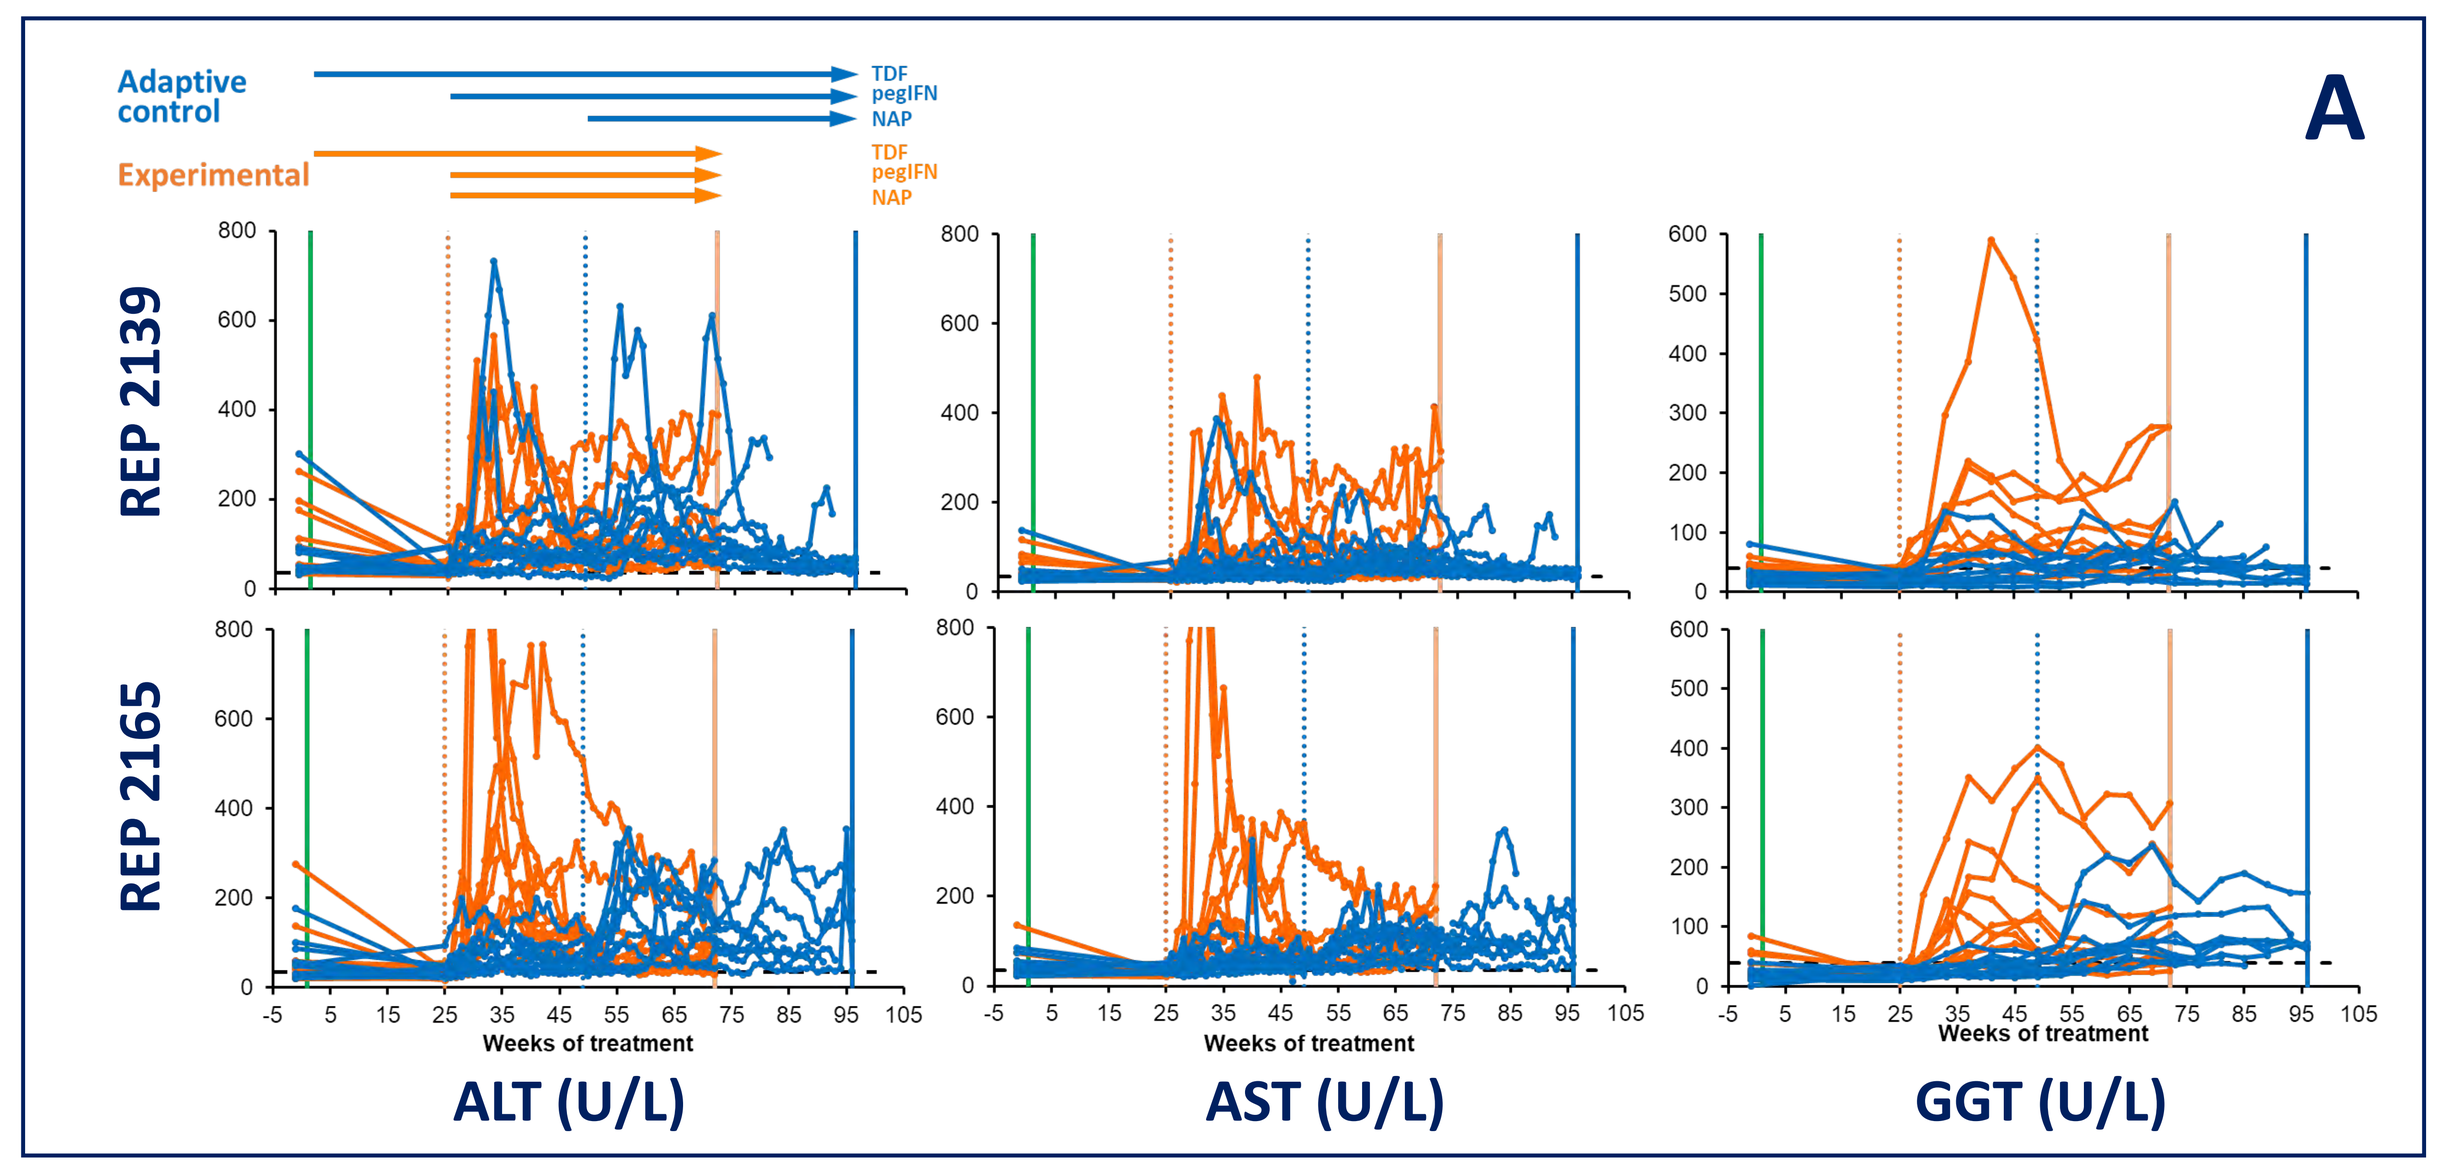
Adaptive control
Experimental
TDF
pegIFN
NAP
A
REP 2139
REP 2165
Weeks of treatment
ALT (U/L)
AST (U/L)
GGT (U/L)

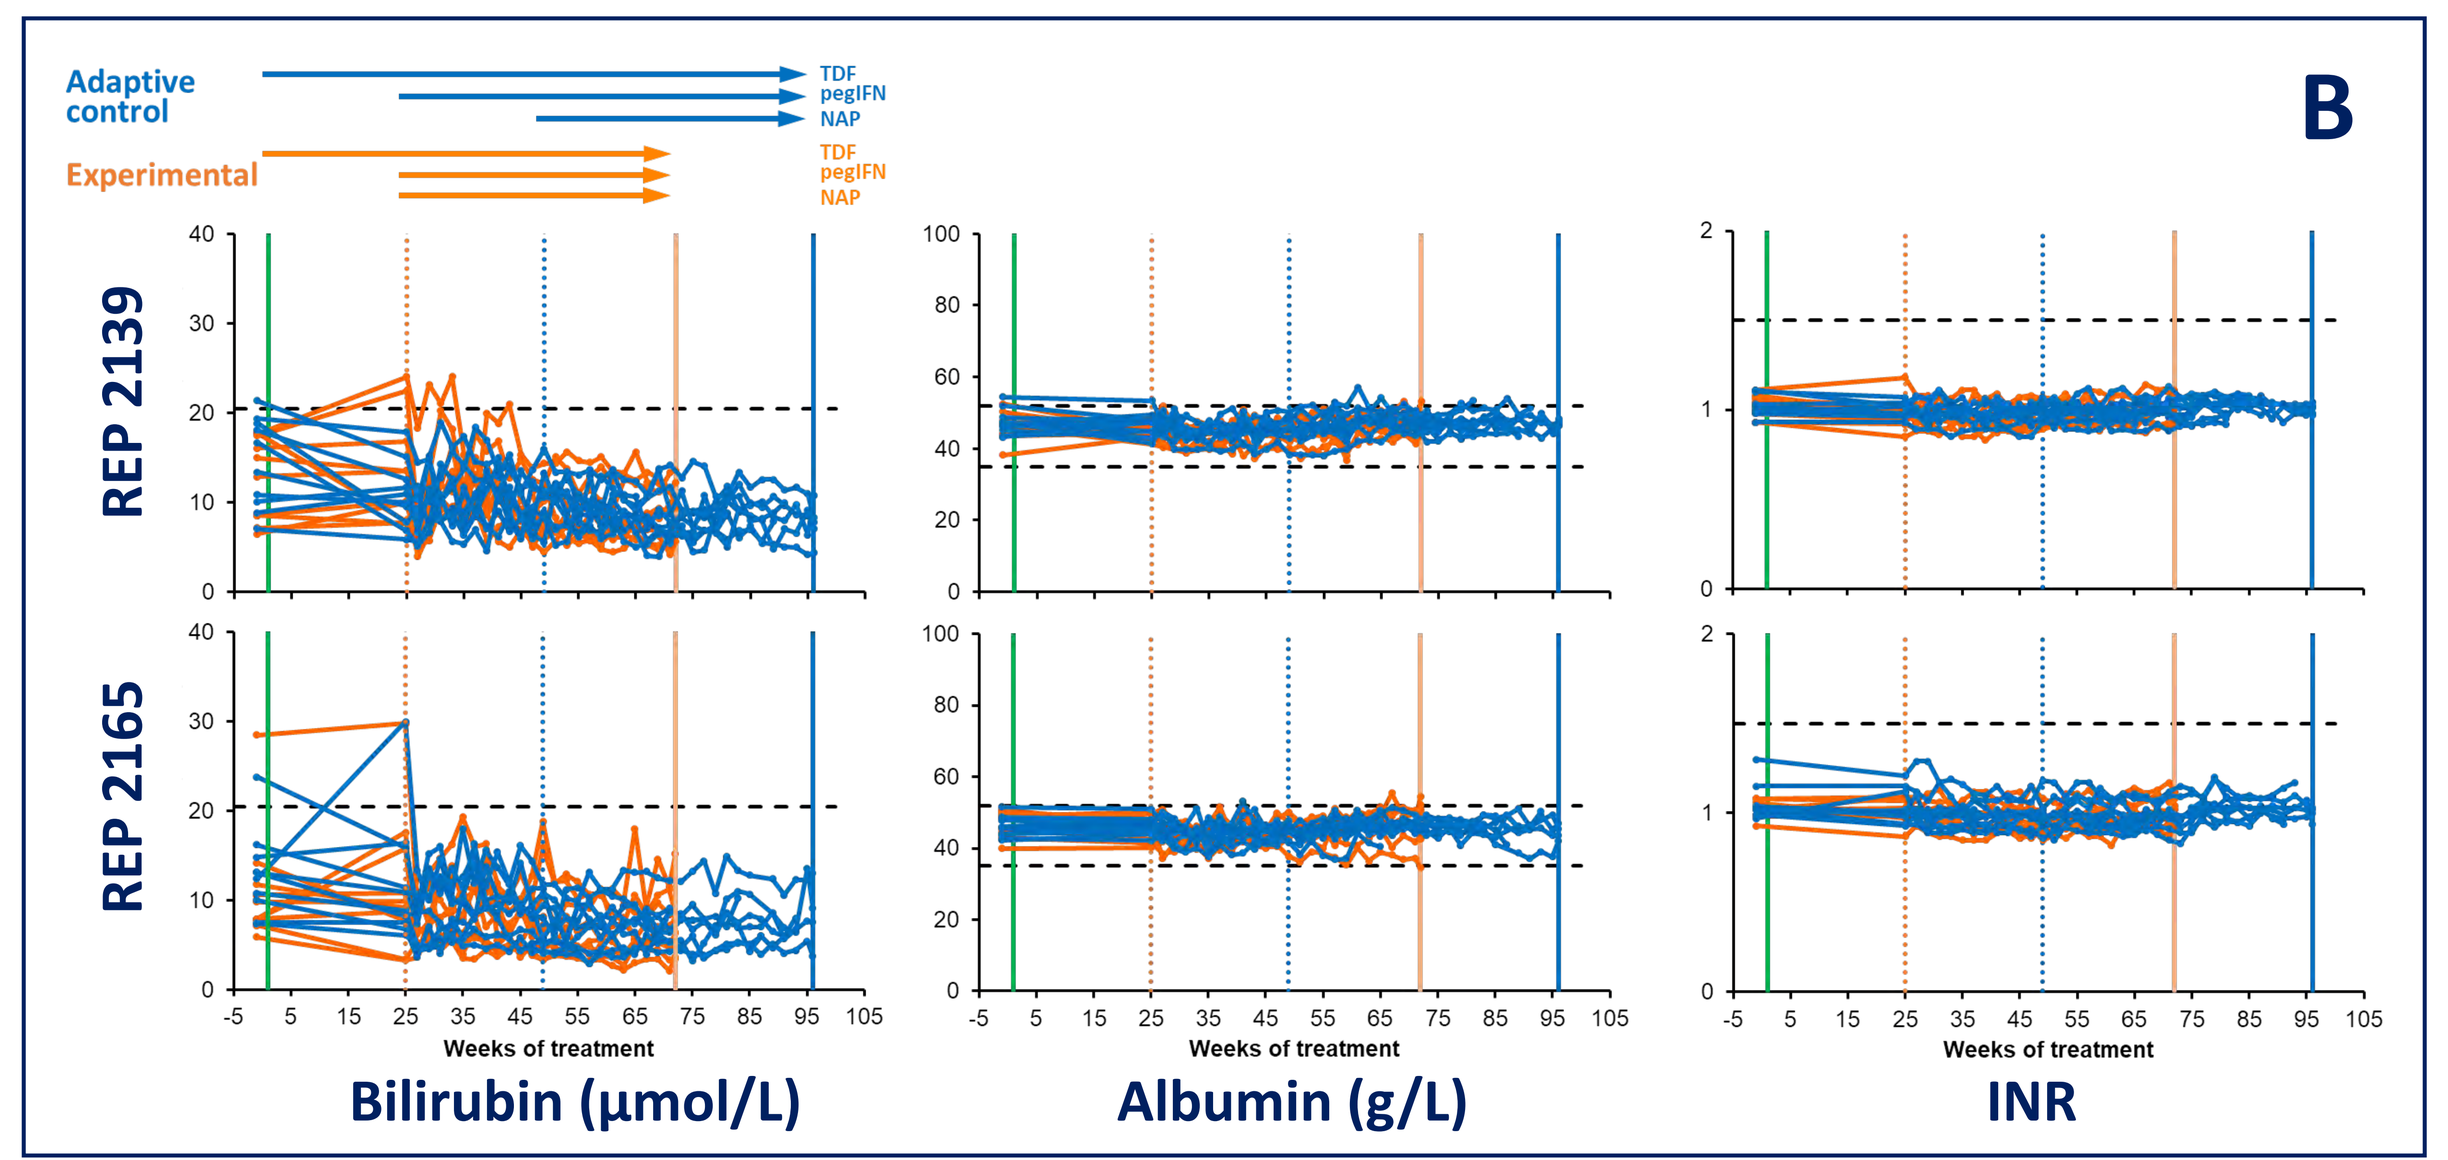
Adaptive control
Experimental
TDF
pegIFN
NAP
B
REP 2139
REP 2165
Weeks of treatment
Bilirubin (µmol/L)
Albumin (g/L)
INR

Liver function

Individual tracings for serum transaminases (A) and liver synthetic function (B) during treatment in the adaptive control (blue) and experimental (orange) groups are provided for patients receiving REP 2139 (top row) and REP 2165 (bottom row). Dashed horizontal lines indicate the upper limit of normal (ALT, AST, GGT, bilirubin, INR) or the normal range (albumin). Liver synthetic functions are normal during all transaminase flares observed during treatment in all patients.

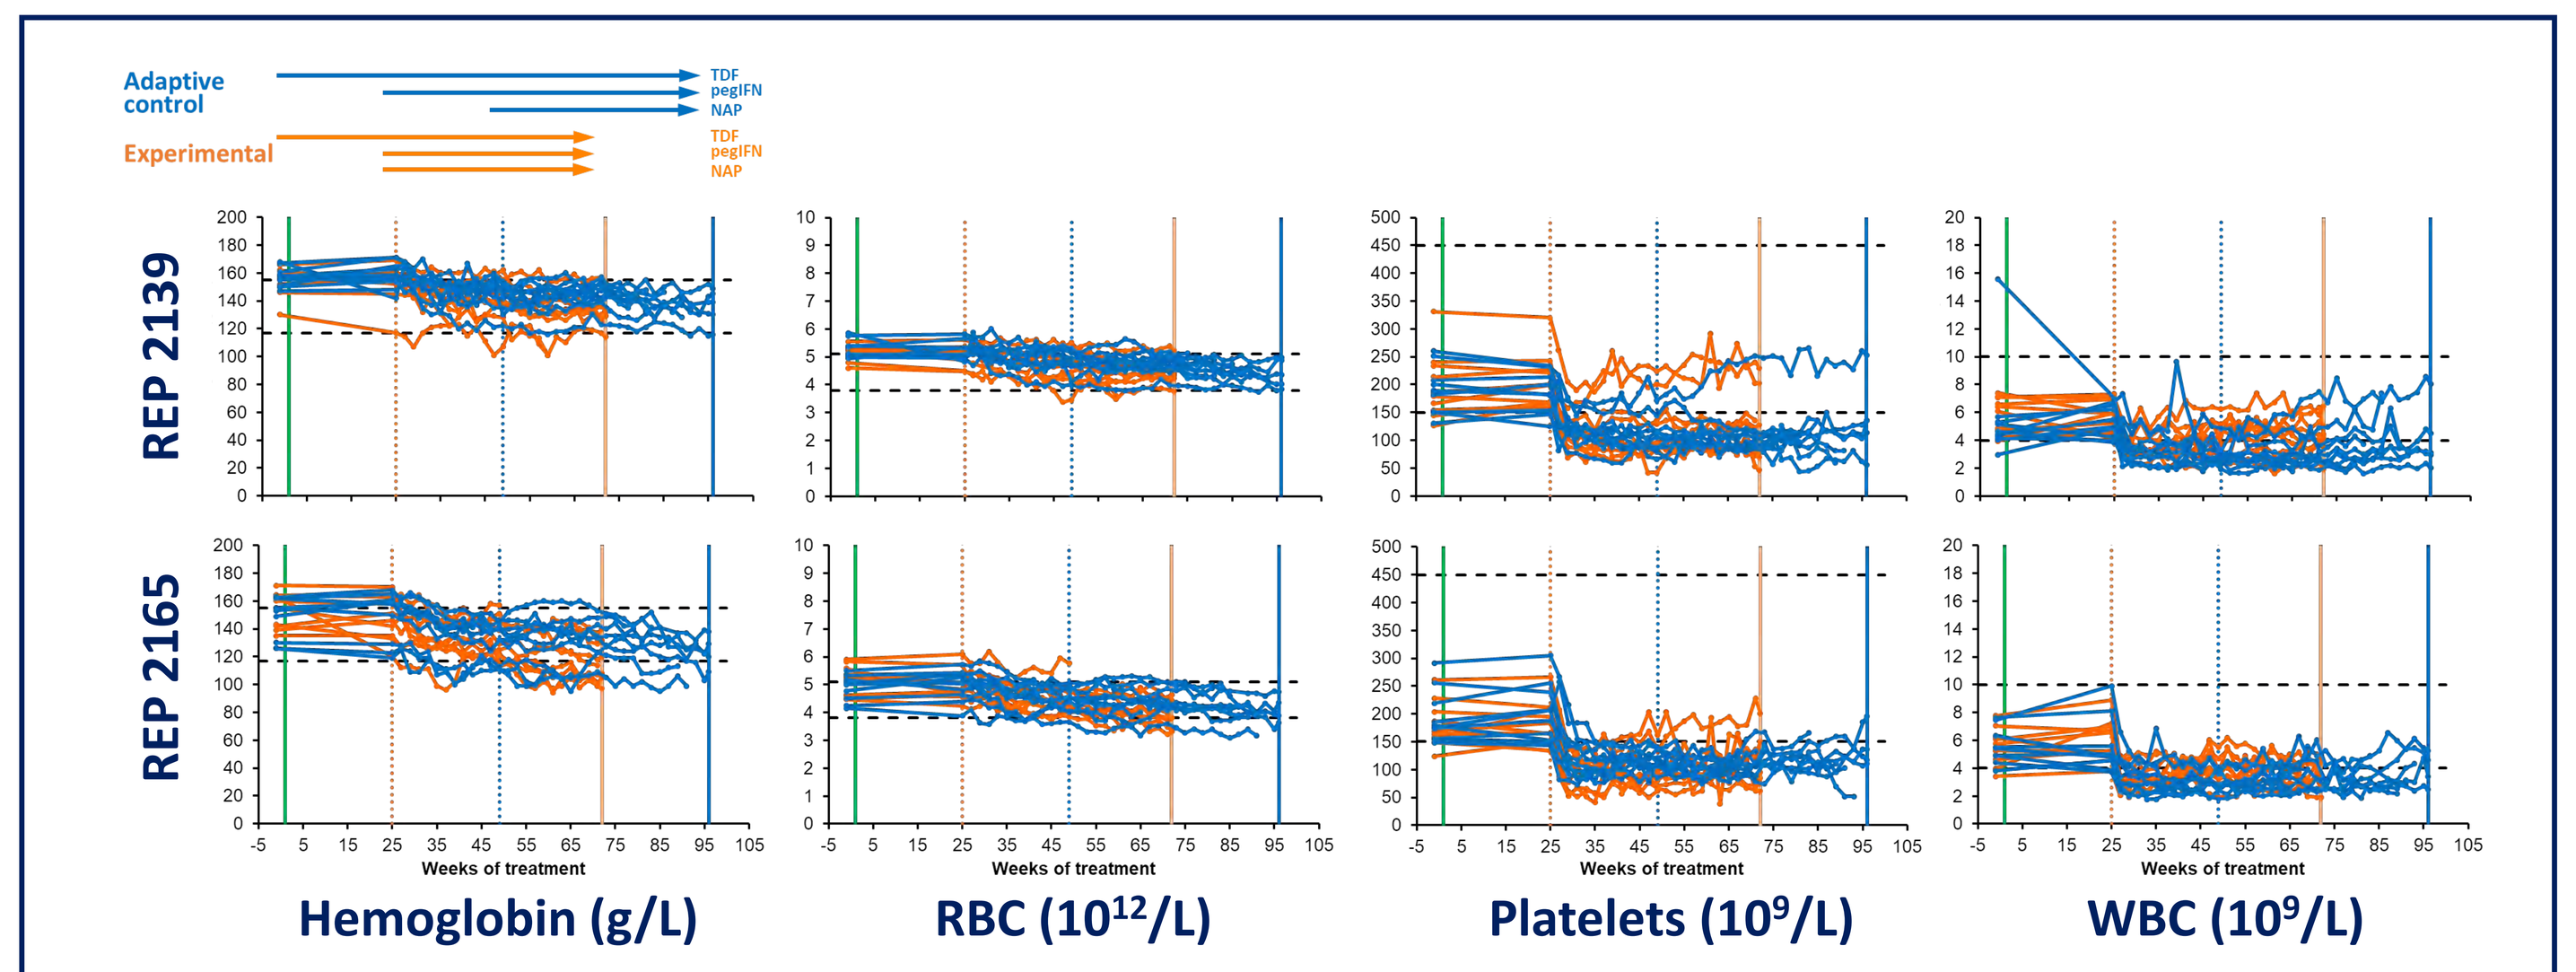
Adaptive control
Experimental
TDF
pegIFN
NAP
REP 2139
REP 2165
Weeks of treatment

Hemoglobin (g/L) RBC (10^{12}/L) Platelets (10^9/L) WBC (10^9/L)

Hematology

Individual hematological analyses during treatment in the adaptive control (blue) and experimental (orange) groups are provided for patients receiving REP 2139 (top row) and REP 2165 (bottom row). Dashed horizontal lines indicate the normal ranges for each parameter. Reductions in platelets and WBC are induced by pegIFN and not impacted by the addition of REP 2139 or REP 2165.

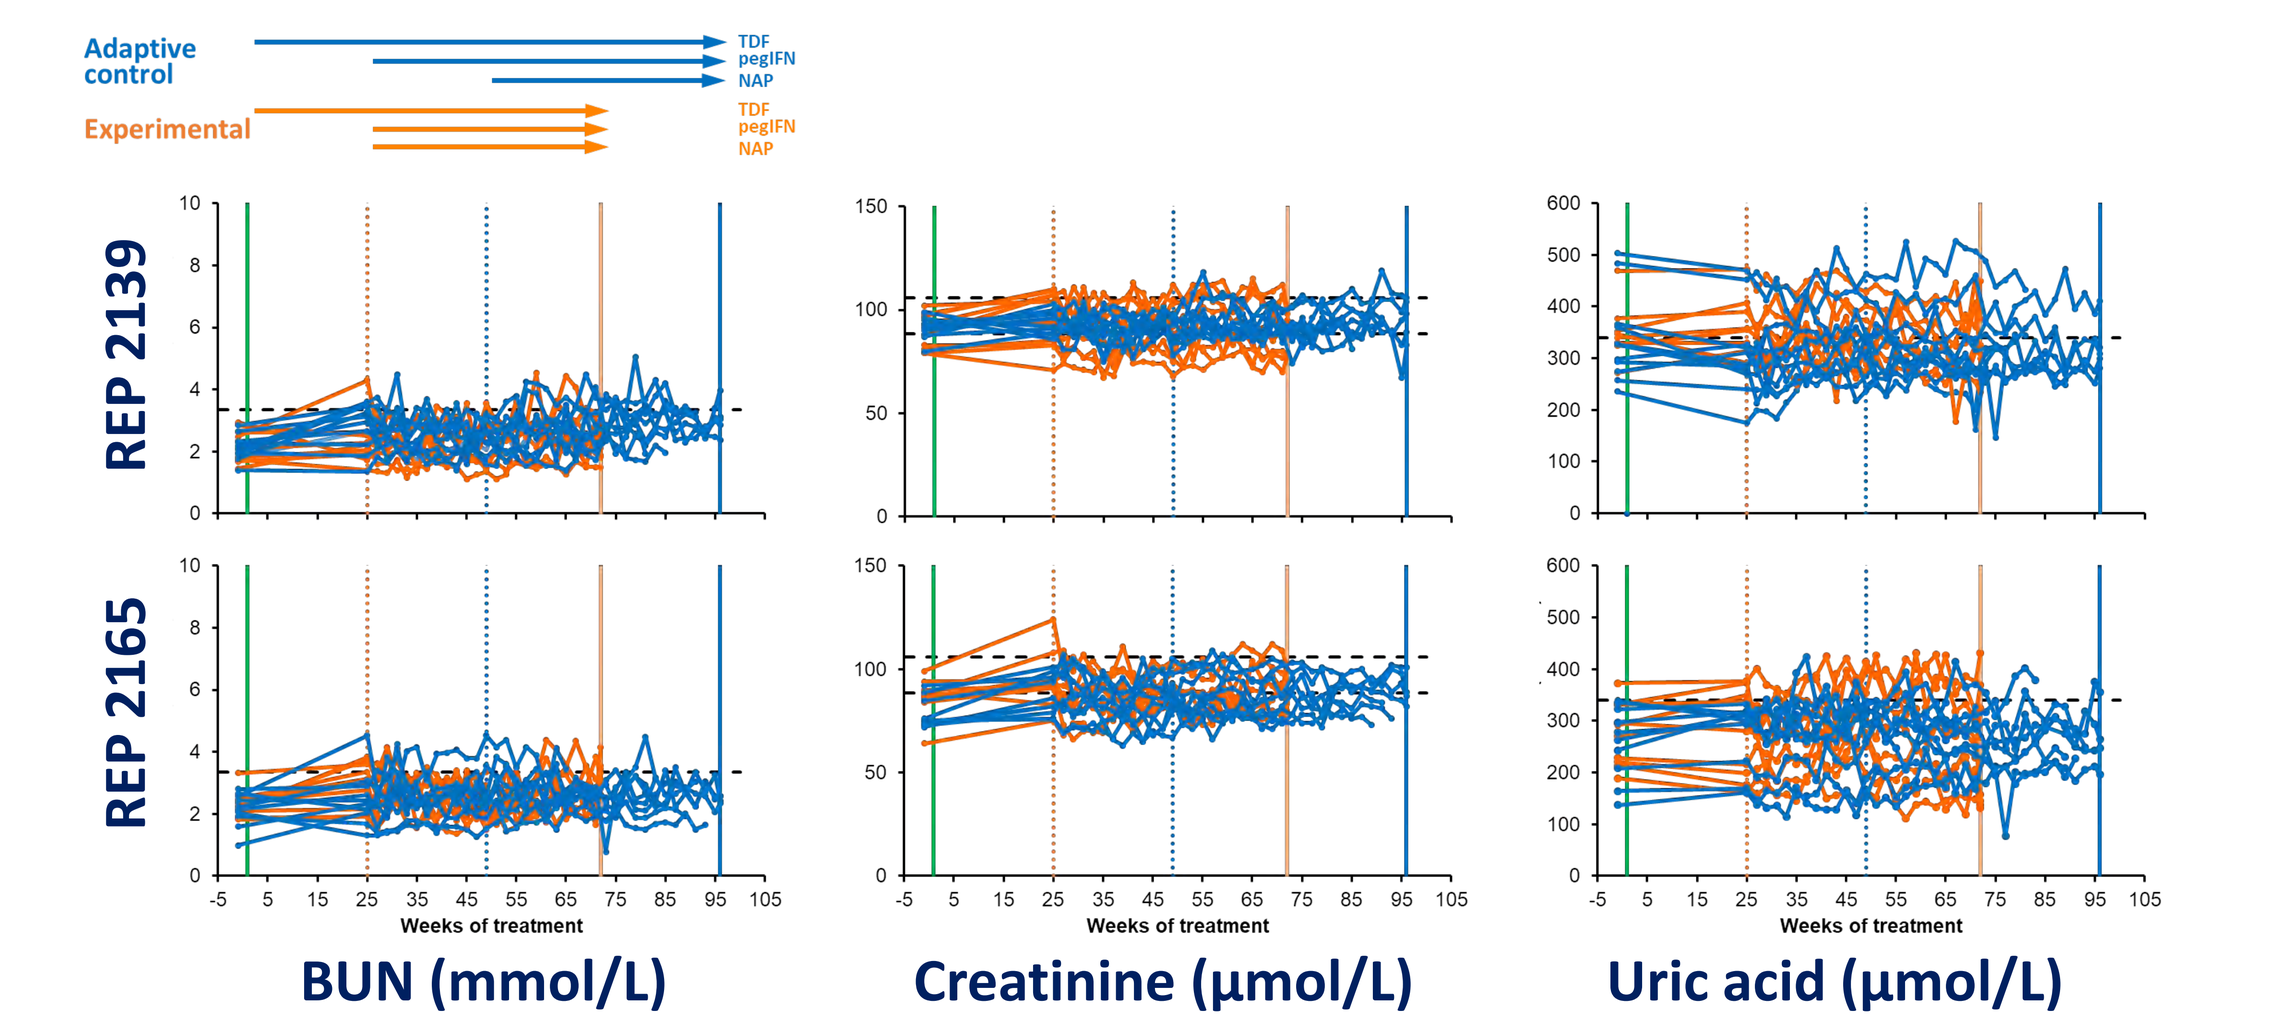
Adaptive control
Experimental
TDF
pegIFN
NAP
REP 2139
REP 2165
Weeks of treatment

BUN (mmol/L) Creatinine (µmol/L) Uric acid (µmol/L)

Renal function

Individual analyses of renal function during treatment in the adaptive control (blue) and experimental (orange) groups are provided for patients receiving REP 2139 (top row) and REP 2165 (bottom row). Dashed horizontal lines indicate the normal ranges for each parameter. Renal function remained normal throughout treatment.

ACKNOWLEDGEMENTS

The authors would like to acknowledge Tanya Korogodina and Dr. Sergiu Merioara for their assistance in supporting the REP 401 trial.

Contact: availlant@replicor.com

REFERENCES

1. Al-Mahtab et al., PLOS ONE 2016;11:e0156667
2. Bazinet et al., Lancet Gastroenterol Hepatol. 2017 epub Sept 27, 2017

INTERIM RESULTS (FOLLOW-UP)

REP 2139 experimental group (Lead clinical candidate)

Patient	Genotype	Parameter	Baseline	EOT	FW4	FW12	FW24
01-003	D1	HBsAg (IU/mL)	7809	TND	TND	TND	TND
		anti-HBs (mIU/mL)	0.61	6147	6287	2631	520
		HBV DNA (IU/ mL)	116900	48	10	TND	TND
		ALT (U/L)	32	115	47	33	26
		AST (U/L)	28	130	53	35	30
02-005	A2	HBsAg (IU/mL)	31184	TND	TND	TND	TND
		anti-HBs (mIU/mL)	<0.1	920	25419	6551	590
		HBV DNA (IU/ mL)	2641	30	<LLOQ	<LLOQ	<LLOQ
		ALT (U/L)	39	304	247	80	45
		AST (U/L)	29	292	179	79	45
02-015	D1	HBsAg (IU/mL)	10921	3510	4256	4807	4520
		anti-HBs (mIU/mL)	<0.1	0.62	0.14	2.84	4.72
		HBV DNA (IU/ mL)	43650	15	74	521900	175
		ALT (U/L)	54	50	19	125	19
		AST (U/L)	30	68	27	79	27
02-024	D1	HBsAg (IU/mL)	1368	TND	TND	TND	TND
		anti-HBs (mIU/mL)	0.64	1.4	1.12	9.14	11
		HBV DNA (IU/ mL)	8312	TND	TND	<LLOQ	<LLOQ
		ALT (U/L)	88	58	25	34	23
		AST (U/L)	38	39	32	28	28
01-046	D2	HBsAg (IU/mL)	8514	TND	TND	TND	<LLOQ*
		anti-HBs (mIU/mL)	<0.1	15529	14037	3404	358*
		HBV DNA (IU/ mL)	70600000	TND	TND	TND	TND*
		ALT (U/L)	112	388	244	37	27
		AST (U/L)	66	315	154	48	37
01-067	D1	HBsAg (IU/mL)	2716	0.31	0.12	0.3	
		anti-HBs (mIU/mL)	<0.1	4.04	1.58	0.54	
		HBV DNA (IU/ mL)	194000	TND	TND	14	
		ALT (U/L)	95	45	39	34	
		AST (U/L)	44	37	29	26	
01-026	D1	HBsAg (IU/mL)	8766	TND	TND	TND	
		anti-HBs (mIU/mL)	<0.1	25	26	16	
		HBV DNA (IU/ mL)	7533000	TND	TND	TND	
		ALT (U/L)	197	124	25	24	
		AST (U/L)	117	103	30	31	
02-019	D1	HBsAg (IU/mL)	9721	TND	TND	TND	
		anti-HBs (mIU/mL)	<0.1	223055	286756	177925	
		HBV DNA (IU/ mL)	4916	17	12	TND	
		ALT (U/L)	41	105	55	36	
		AST (U/L)	31	63	37	30	
01-077	D2	HBsAg (IU/mL)	1334	TND	TND		
		anti-HBs (mIU/mL)	5.81	38	30		
		HBV DNA (IU/ mL)	2183000	TND	TND		
		ALT (U/L)	263	70	34	24	
		AST (U/L)	78	61	33	25	
03-023	D1	HBsAg (IU/mL)	9595	(3006)			
		anti-HBs (mIU/mL)	12	(0.79)			
		HBV DNA (IU/ mL)	1132000	(TND)			
		ALT (U/L)	175	(39)			
		AST (U/L)	85	(34)			

*updated data not available for e-poster

REP 2165 experimental group

Patient	Genotype	Parameter	Baseline	EOT	FW4	FW12	FW24
01-004	D3	HBsAg (IU/mL)	9032	238	240	166	
		anti-HBs (mIU/mL)	<0.1	<0.1	<0.1	<0.1	
		HBV DNA (IU/ mL)	3020	1	10	26320	
		ALT (U/L)	59	227	138	41	
		AST (U/L)	38	172	82	54	
01-007	D1	HBsAg (IU/mL)	9418	TND	TND	TND	<LLOQ
		anti-HBs (mIU/mL)	<0.1	331	345	179	44
		HBV DNA (IU/ mL)	13300	TND	<LLOQ	<LLOQ	TND
		ALT (U/L)	41	82	38	22	17
		AST (U/L)	24	62	35	25	19
01-010	D2	HBsAg (IU/mL)	14498	1388	1372	1865	9852
		anti-HBs (mIU/mL)	<0.1	<0.1	<0.1	<0.1	<0.1
		HBV DNA (IU/ mL)	14190000	10	81	1612000	6102000
		ALT (U/L)	276	39	20	32	176
		AST (U/L)	136	47	27	31	70
01-042	D1	HBsAg (IU/mL)	3049	TND	TND	TND	TND
		anti-HBs (mIU/mL)	<0.1	94	69	20	19
		HBV DNA (IU/ mL)	53080	TND	TND	TND	TND
		ALT (U/L)	44	229	90	66	43
		AST (U/L)	27	222	72	48	38
02-049	D2	HBsAg (IU/mL)	1147	TND	TND	TND	
		anti-HBs (mIU/mL)	<0.1	547	221	172	
		HBV DNA (IU/ mL)	164900	<LLOQ	TND	TND	
		ALT (U/L)	59	28	17	14	
		AST (U/L)	55	38	31	26	
01-008	D2	HBsAg (IU/mL)	10736	57.9	87.1	1013	
		anti-HBs (mIU/mL)	35	1.5	1.22	1.94	
		HBV DNA (IU/ mL)	236300	10	15	51910	
		ALT (U/L)	137	54	40	46	
		AST (U/L)	74	49	38	44	
02-054	D1	HBsAg (IU/mL)	18422	TND	TND	TND	
		anti-HBs (mIU/mL)	<0.1	36628	43489	31150	
		HBV DNA (IU/ mL)	2130	TND	TND	TND	
		ALT (U/L)	24	62	27	18	
		AST (U/L)	24	61	36	30	
02-058	A2	HBsAg (IU/mL)	8720	TND	TND		
		anti-HBs (mIU/mL)	<0.1	13582	16366		
		HBV DNA (IU/ mL)	33000	TND	TND		
		ALT (U/L)	32	83	45		
		AST (U/L)	46	77	56		
01-074	D2	HBsAg (IU/mL)	4835	631	701		
		anti-HBs (mIU/mL)	<0.1	<0.1	<0.1		
		HBV DNA (IU/ mL)	14550	10	80		
		ALT (U/L)	20	73	28		
		AST (U/L)	24	66	34		
03-020	D2	HBsAg (IU/mL)	8577	(TND)			
		anti-HBs (mIU/mL)	<0.1	(148)			
		HBV DNA (IU/ mL)	38530	(26)			
		ALT (U/L)	51	(155)			
		AST (U/L)	41	(100)			

HBsAg reduction to <1IU/mL during treatment **>1log_{10} HBsAg reduction but >1IU/mL** **Functional control after removal of therapy**

TND = target not detected (HBsAg <0.01IU/mL, HBV DNA no target detected), LLOQ = lower limit of quantification (HBsAg <0.05IU/mL, HBV DNA <10IU/mL)

Numbers in parentheses indicate the latest available on-treatment results for patients who have not completed treatment.

Persistence of functional control after therapy

Available baseline, end of treatment and follow-up data for HBsAg, anti-HBs, HBV DNA, ALT and AST are provided for all patients in the REP 2139 and REP 2165 experimental groups. Persistence of functional control of HBV infection and normalization of ALT and AST (ULN = 50 U/L) are present in 8/10 patients in the REP 2139 group and 5/10 patients in the REP 2165 group. Three patients in the REP 2165 group (in orange) experienced >1 log_{10} reductions in serum HBsAg but remained >1 IU/mL the end of therapy and experienced viral rebound after withdrawal of therapy. Recovery of platelet and WBC counts are observed at 12 weeks of follow-up in all experimental patients (inset right). Dashed horizontal lines indicate normal ranges.

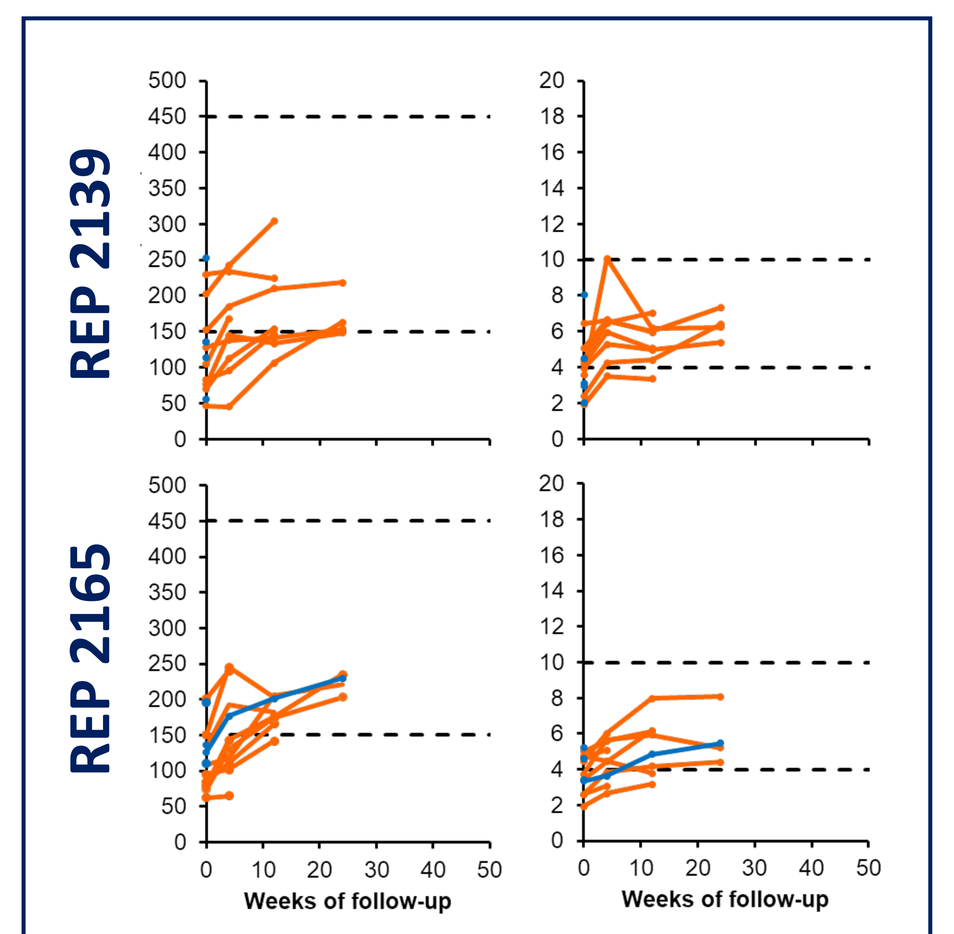
REP 2139
REP 2165
Weeks of follow-up

Platelets (10^9/L) WBC (10^9/L)

CONCLUSIONS

- **REP 2139 and REP 2165 are well tolerated in combination with TDF and pegIFN.**
- **Clearance of HBsAg uniquely occurs with NAP exposure**
- **In the presence of pegIFN, HBsAg clearance is accompanied by dramatic increases in circulating anti-HBs and the increased prevalence and magnitude of otherwise asymptomatic transaminase flares.**
- **Functional control of infection is persisting after therapy in all experimental patients achieving HBsAg <1 IU/mL during treatment:**

 8/10 with REP 2139
 5/10 with REP 2165

- **All patients with functional control of infection persisting after therapy have also experienced normalization of liver enzymes.**