REP 2139 targets the Hepatitis Delta Virus (HDV) ribonucleoprotein (RNP) and exerts a direct antiviral effect on HDV replication

Simone FONTE [1,2], Marie Laure PLISSONNIER [1,2], Maud MICHELET [1,2], Matthieu BLANCHET [3,4], Patrick LABONTE' [4], Andrew VAILLANT [3], Massimo LEVRERO [1,2,5]

[1] Hepatology Institute, Lyon, France; [2] Cancer Research Center of Lyon (CRCL), INSERM U1052, CNRS UMR5286, Lyon, France; [3] Replicor Inc., Montreal, Canada; [4] INRS – Institut Armand Frappier, Institut National de la Recherche Scientifique, Laval, Canada; [5] Department of Hepatology, Hôpital Croix-Rousse, Hospices Civils de Lyon, Lyon, France; [6] University of Lyon Claude Bernard 1 (UCLB1), Lyon, France

Introduction and objectives

The nucleic acid polymer (NAP) REP 2139 has multiple molecular mechanisms: 1) targeting the HSP40 chaperone DNAJB12 to block HBV subviral particles assembly and secretion (Boulon et al, Hepatology (S1) 2020; Angelo et al, HBV Meeting 2023), an effect which also blocks HDV ribonuclear protein (RNP) envelopment and HDV secretion from infected cell; 2) direct binding to S-HDAg and L-HDAg (Shamur et al, Hepatology (S1) 2017). A phase II study (Bazinet et al, Lancet GastroHep 2017) and current compassionate use of REP 2139 in HBV/ HDV infection have shown a more robust and early response toward HDV RNA as compared to HBsAg, suggesting a second direct acting antiviral mechanism. Here we have investigated the direct REP 2139 antiviral activity in relevant HDV cell infection models in vitro.

Methods

REP 2139 endosomal release in vitro was restored by UNC 7938 (Blanchet, Antiviral Res 2019). Clinical supply of REP 2139-Mg (lot FAB-22-0001) was used for dosing in HDV infected (10 ge/cell) HepG2-NTCP cells and primary human hepatocytes (PHH) and in Huh7-END cells stably replicating HDV (Ni Y, SciRep 2019). Intracellular HDV viral genome levels were assessed by qRT-PCR. HDV RNA and Hepatitis Delta Antigen (HDAg) association to form the HDV ribonucleoprotein (HDV RNP) was monitored by anti-HDAg RNA immunoprecipitation (RIP) followed by HDV qRT-PCR (Abeywickrama-Samarakoon N, Nat Comms 2020).

Results

1. A single dose of REP 2139-Mg reduced intracellular HDV viral genome levels by ~1 log10 in HepG2-NTCP, PHH cells and Huh7-END at 400nM, 600 nM and 600nM respectively.

2. Loss of antiviral activity at higher doses could be recovered by increasing UNC 7938 concentration, indicating that REP 2139 endosomal release is influenced by both REP 2139 endosomal concentration and UNC 7938 dosing.

3. REP 2139-Mg reduces both genomic (≈1Log) and antigenomic (≈1.5Log) HDV RNA.

4. A single REP 2139-Mg dose also reduced the intranuclear association of HDV RNA with HDAg by ~60% (@ 600nM) in HepG2-NTCP and by 65% (@ 400nM), without changing the HDAg protein levels.

5. REP 2139-Mg inhibits HDV replication in HBV-HDV coinfected PHHs. We confirm in HBV-HDV coinfected PHHs that REP 2139 inhibits HBsAg secretion without affecting intracellular pgRNA levels or HBeAg secretion.

REP 2139 putative molecular mechanisms

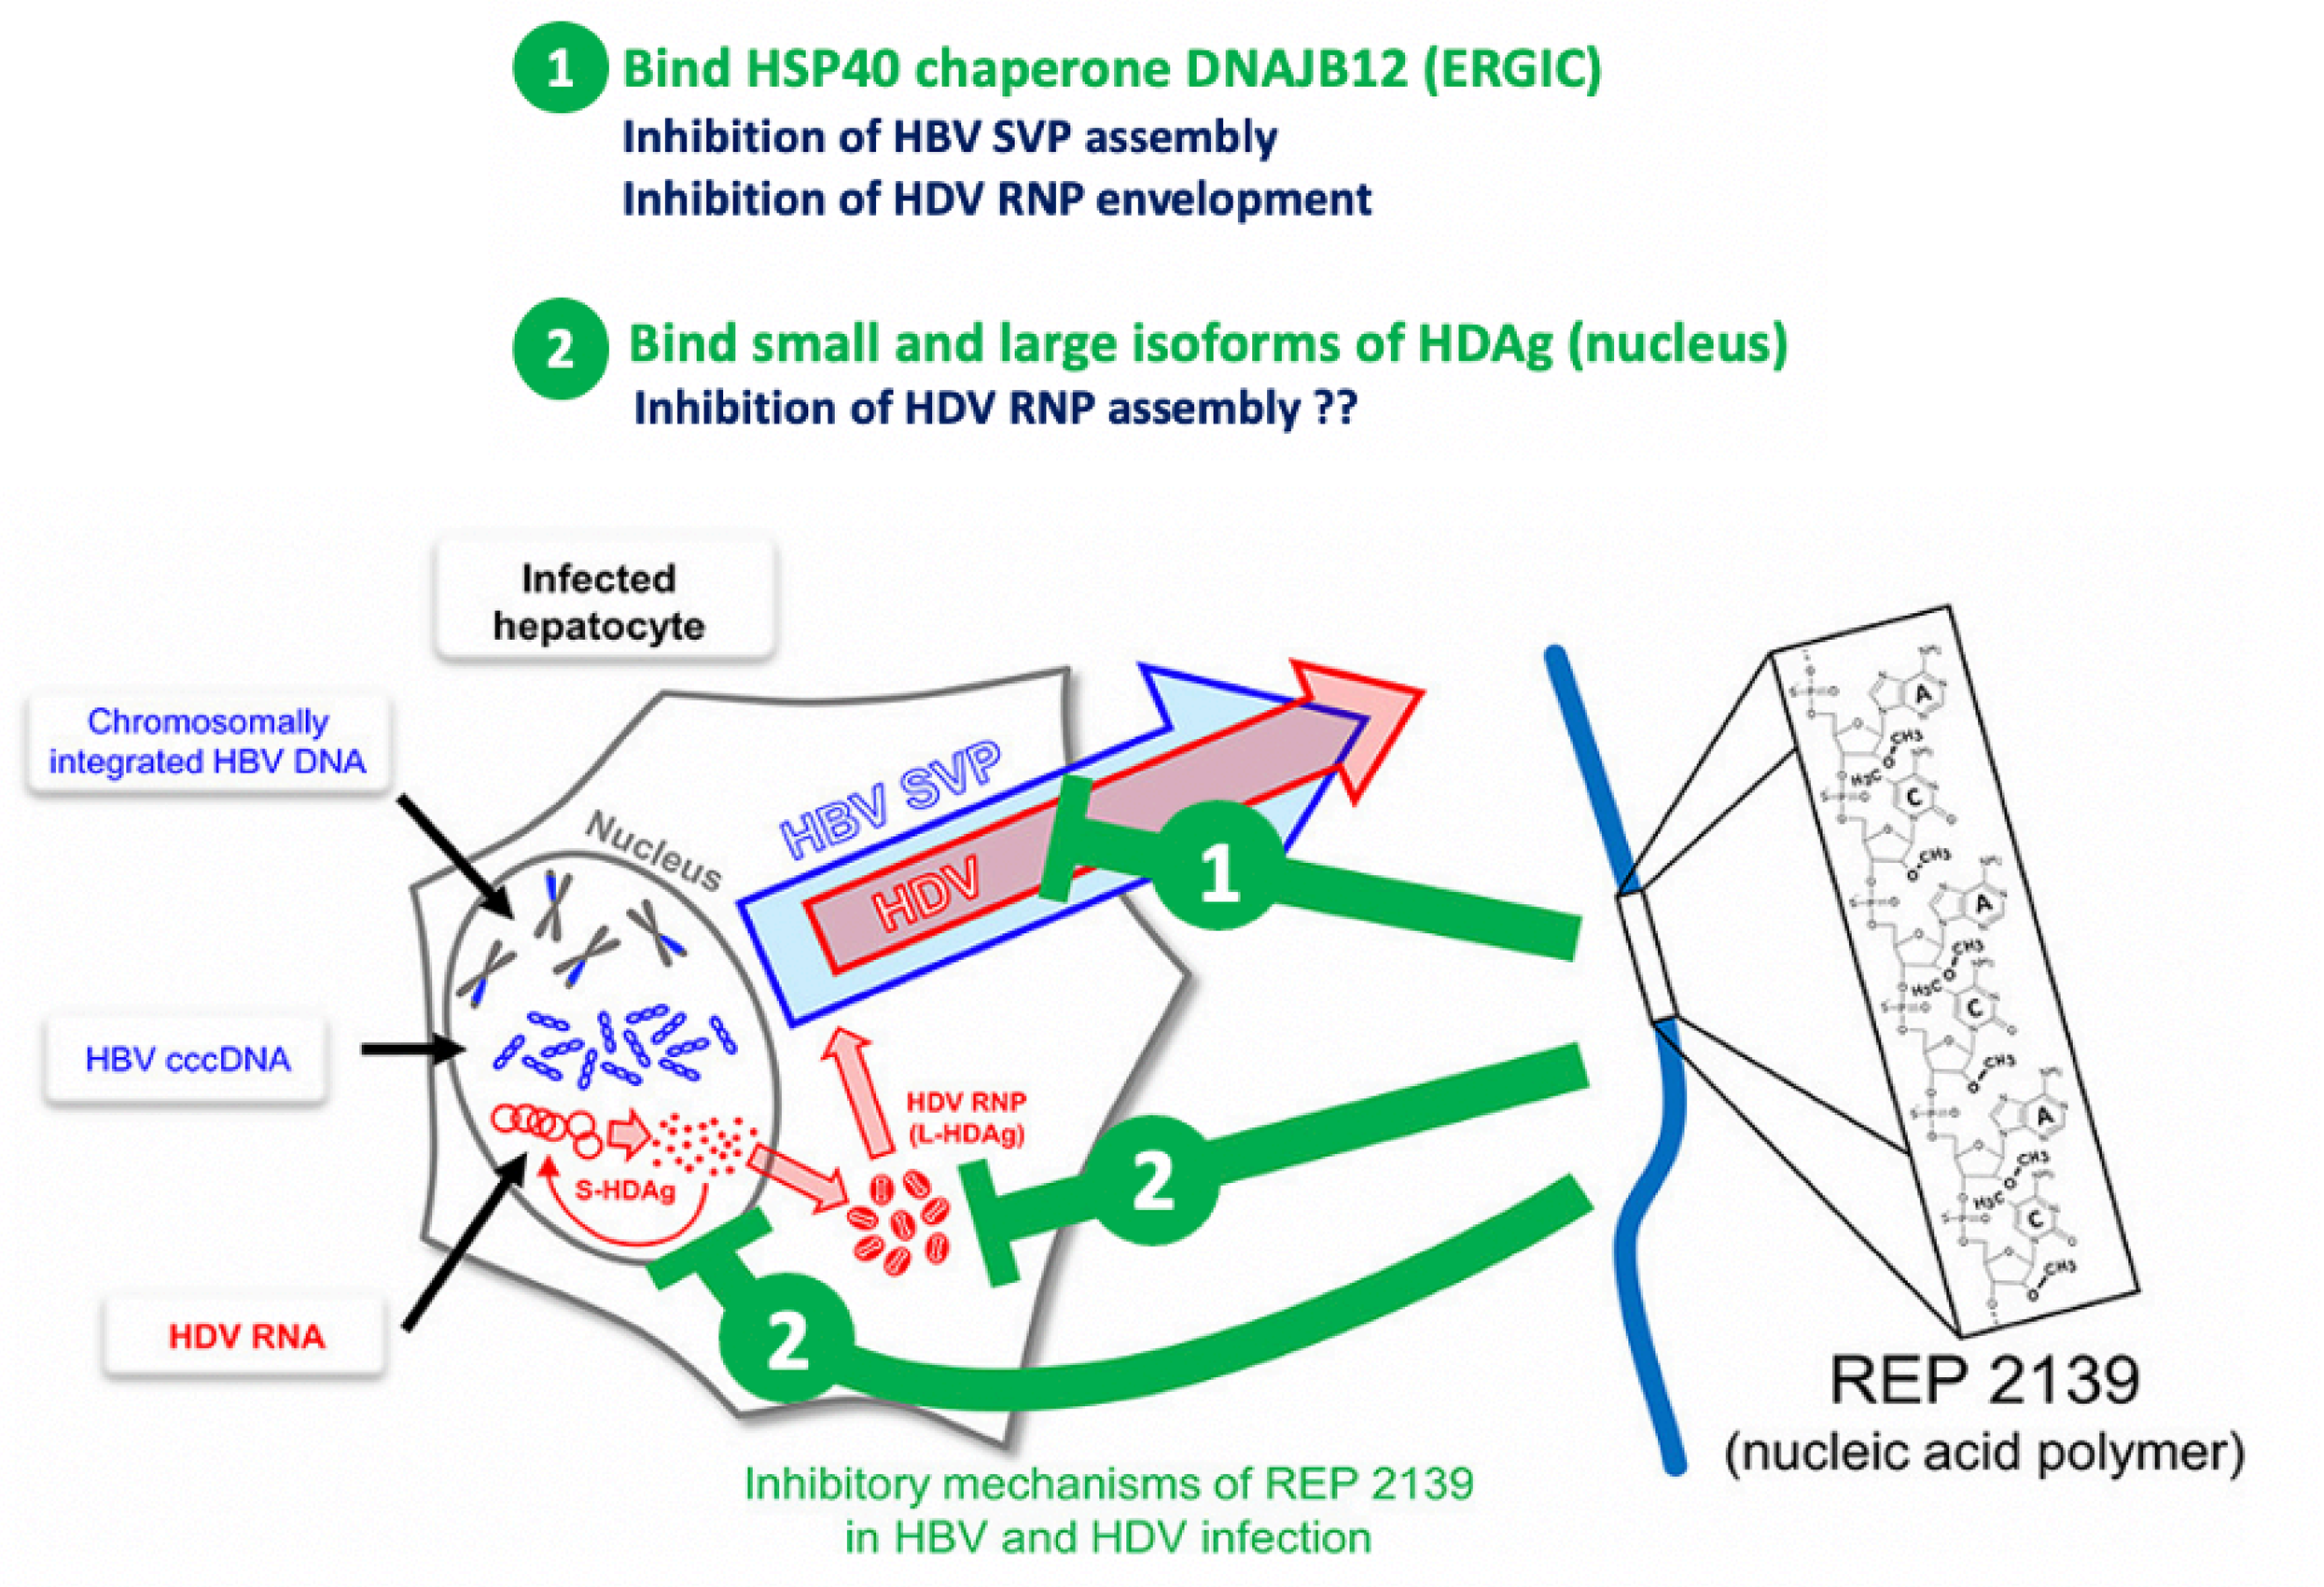
1 Bind HSP40 chaperone DNAJB12 (ERGIC)
Inhibition of HBV SVP assembly
Inhibition of HDV RNP envelopment
2 Bind small and large isoforms of HDAg (nucleus)
Inhibition of HDV RNP assembly ??
Infected hepatocyte
Chromosomally integrated HBV DNA
HBV cccDNA
HDV RNA
Nucleus
HBV SVP
HDV
HDV RNP (L-HDAg)
S-HDAg
REP 2139
(nucleic acid polymer)
Inhibitory mechanisms of REP 2139 in HBV and HDV infection

REP2139 in HDV patients

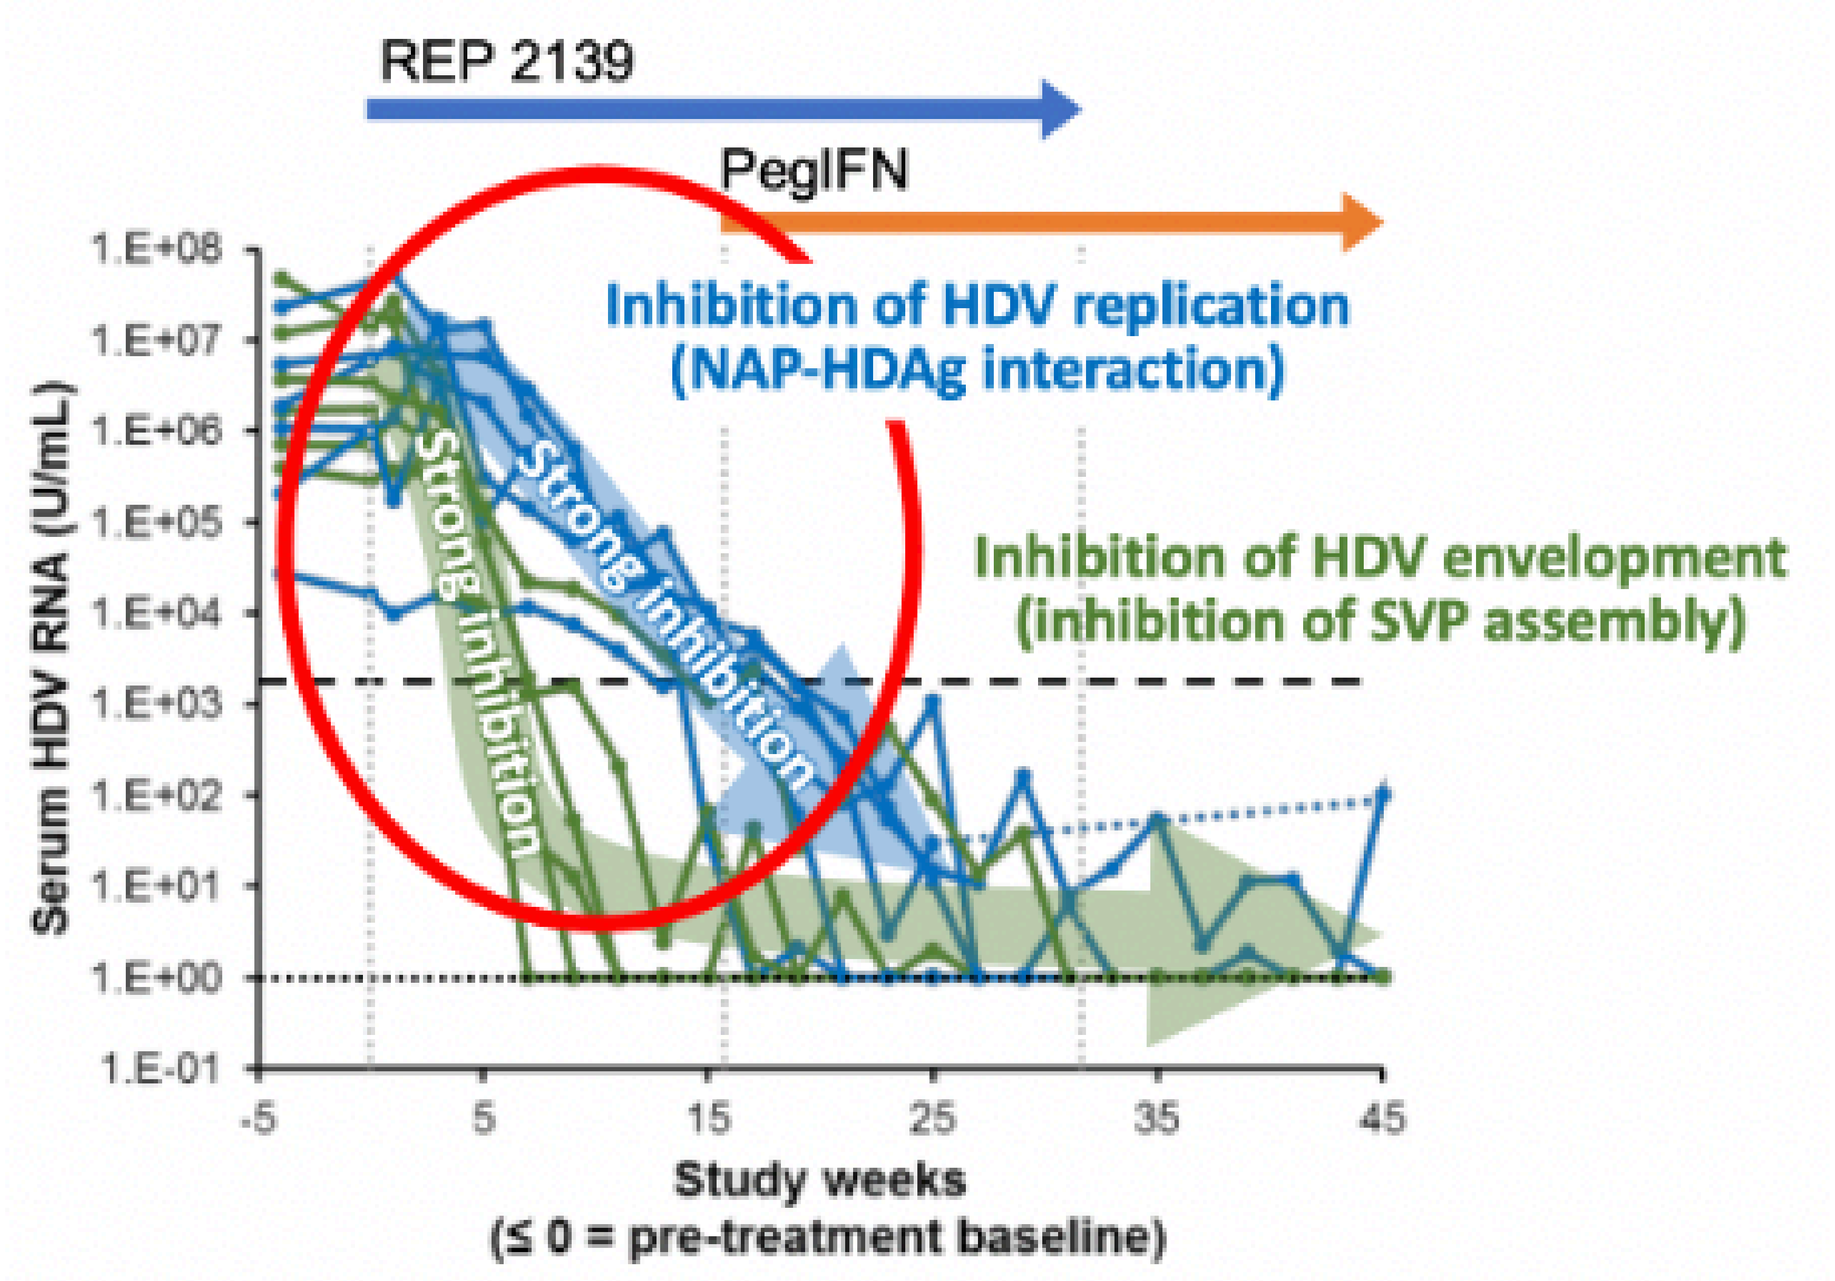
REP 2139
PegIFN
Inhibition of HDV replication (NAP-HDAg interaction)
Inhibition of HDV envelopment (inhibition of SVP assembly)
Strong inhibition
Strong inhibition
Serum HDV RNA (U/mL)
Study weeks
(≤ 0 = pre-treatment baseline)

The effect on HDV RNA before PEG-IFN is started

Conclusions

REP 2139 has a direct acting antiviral effect against HDV RNA replication which may involve blocking HDV RNA interaction with HDAg during HDV RNP morphogenesis. These antiviral effects bear further investigation and may explain the more rapid decline of HDV RNA versus HBsAg in human studies.

Results 1.1 A single dose of REP 2139-Mg reduces intracellular HDV viral genome levels by ~1 $\log_{10}$ in HepG2-NTCP and PHH cells

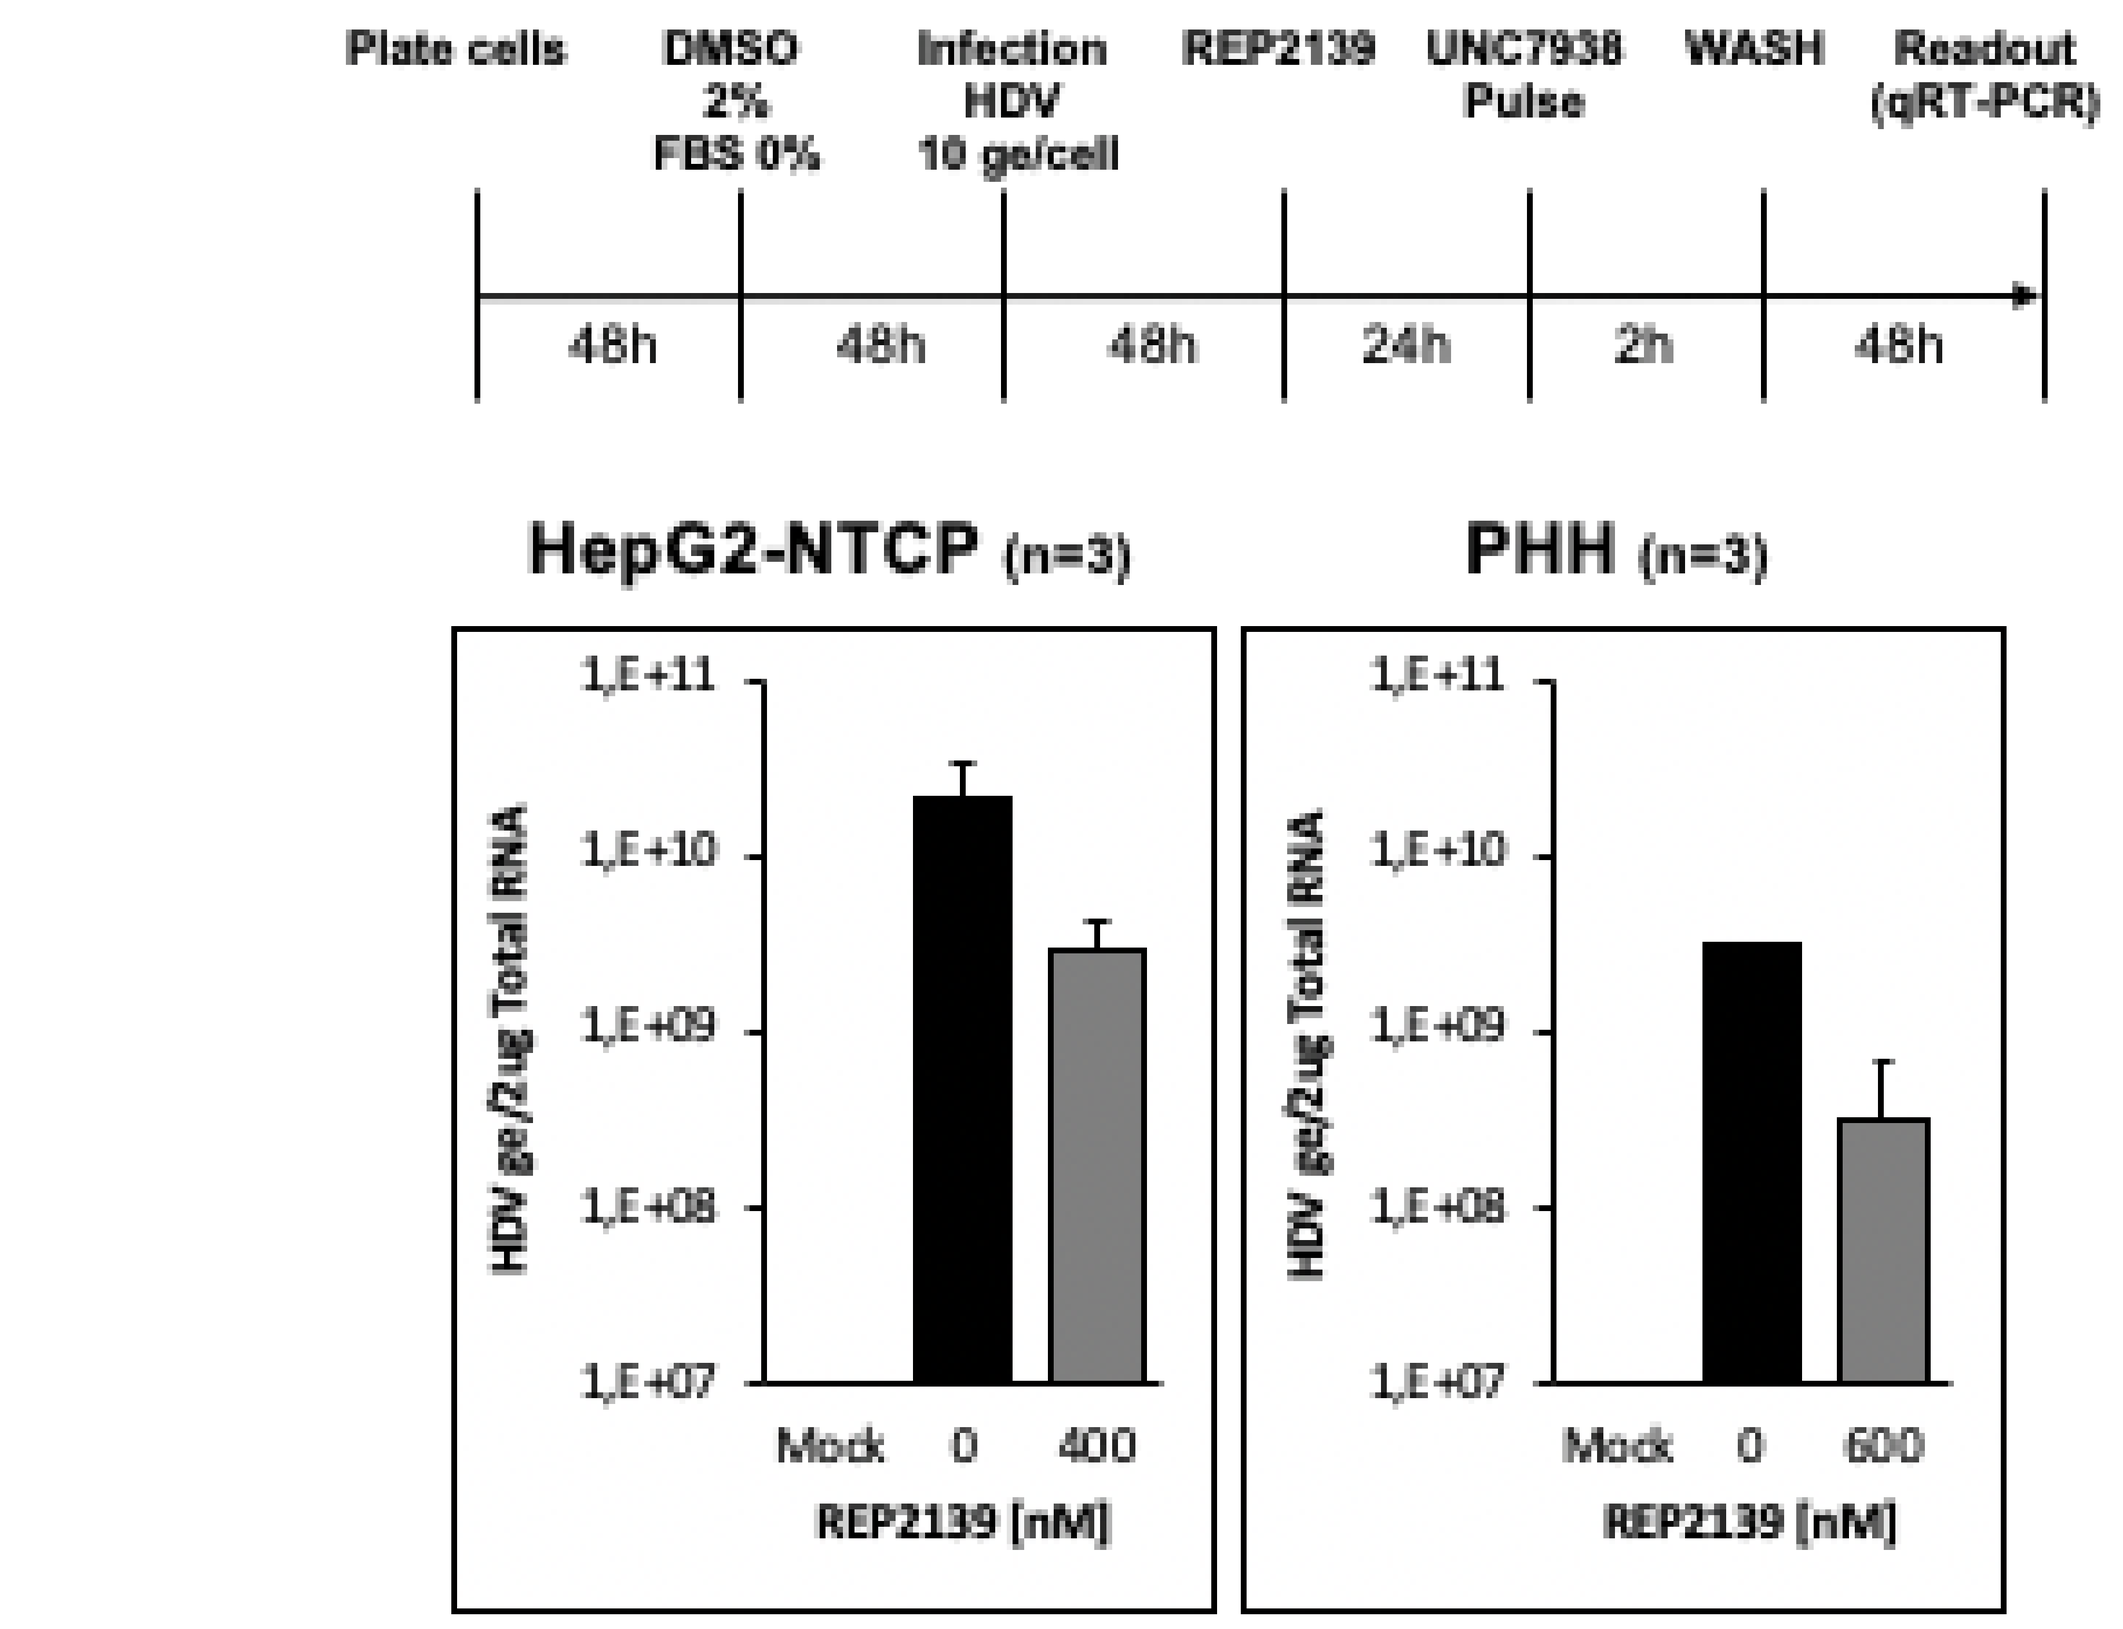
Plate cells
DMSO 2% FBS 0%
Infection HDV 10 ge/cell
REP2139
UNC7938 Pulse
WASH
Readout (qRT-PCR)
48h 48h 48h 24h 2h 48h
HepG2-NTCP (n=3)
PHH (n=3)
HDV ge/2ug Total RNA
Mock 0 400
Mock 0 600
REP2139 [nM]

Results 1.2 A single dose of REP 2139-Mg reduces intracellular HDV viral genome levels by ~1 $\log_{10}$ also in the stable HBD replicating cells Huh7-END cell line that supports a complete HDV viral cycle and secretes HDV infectious virions

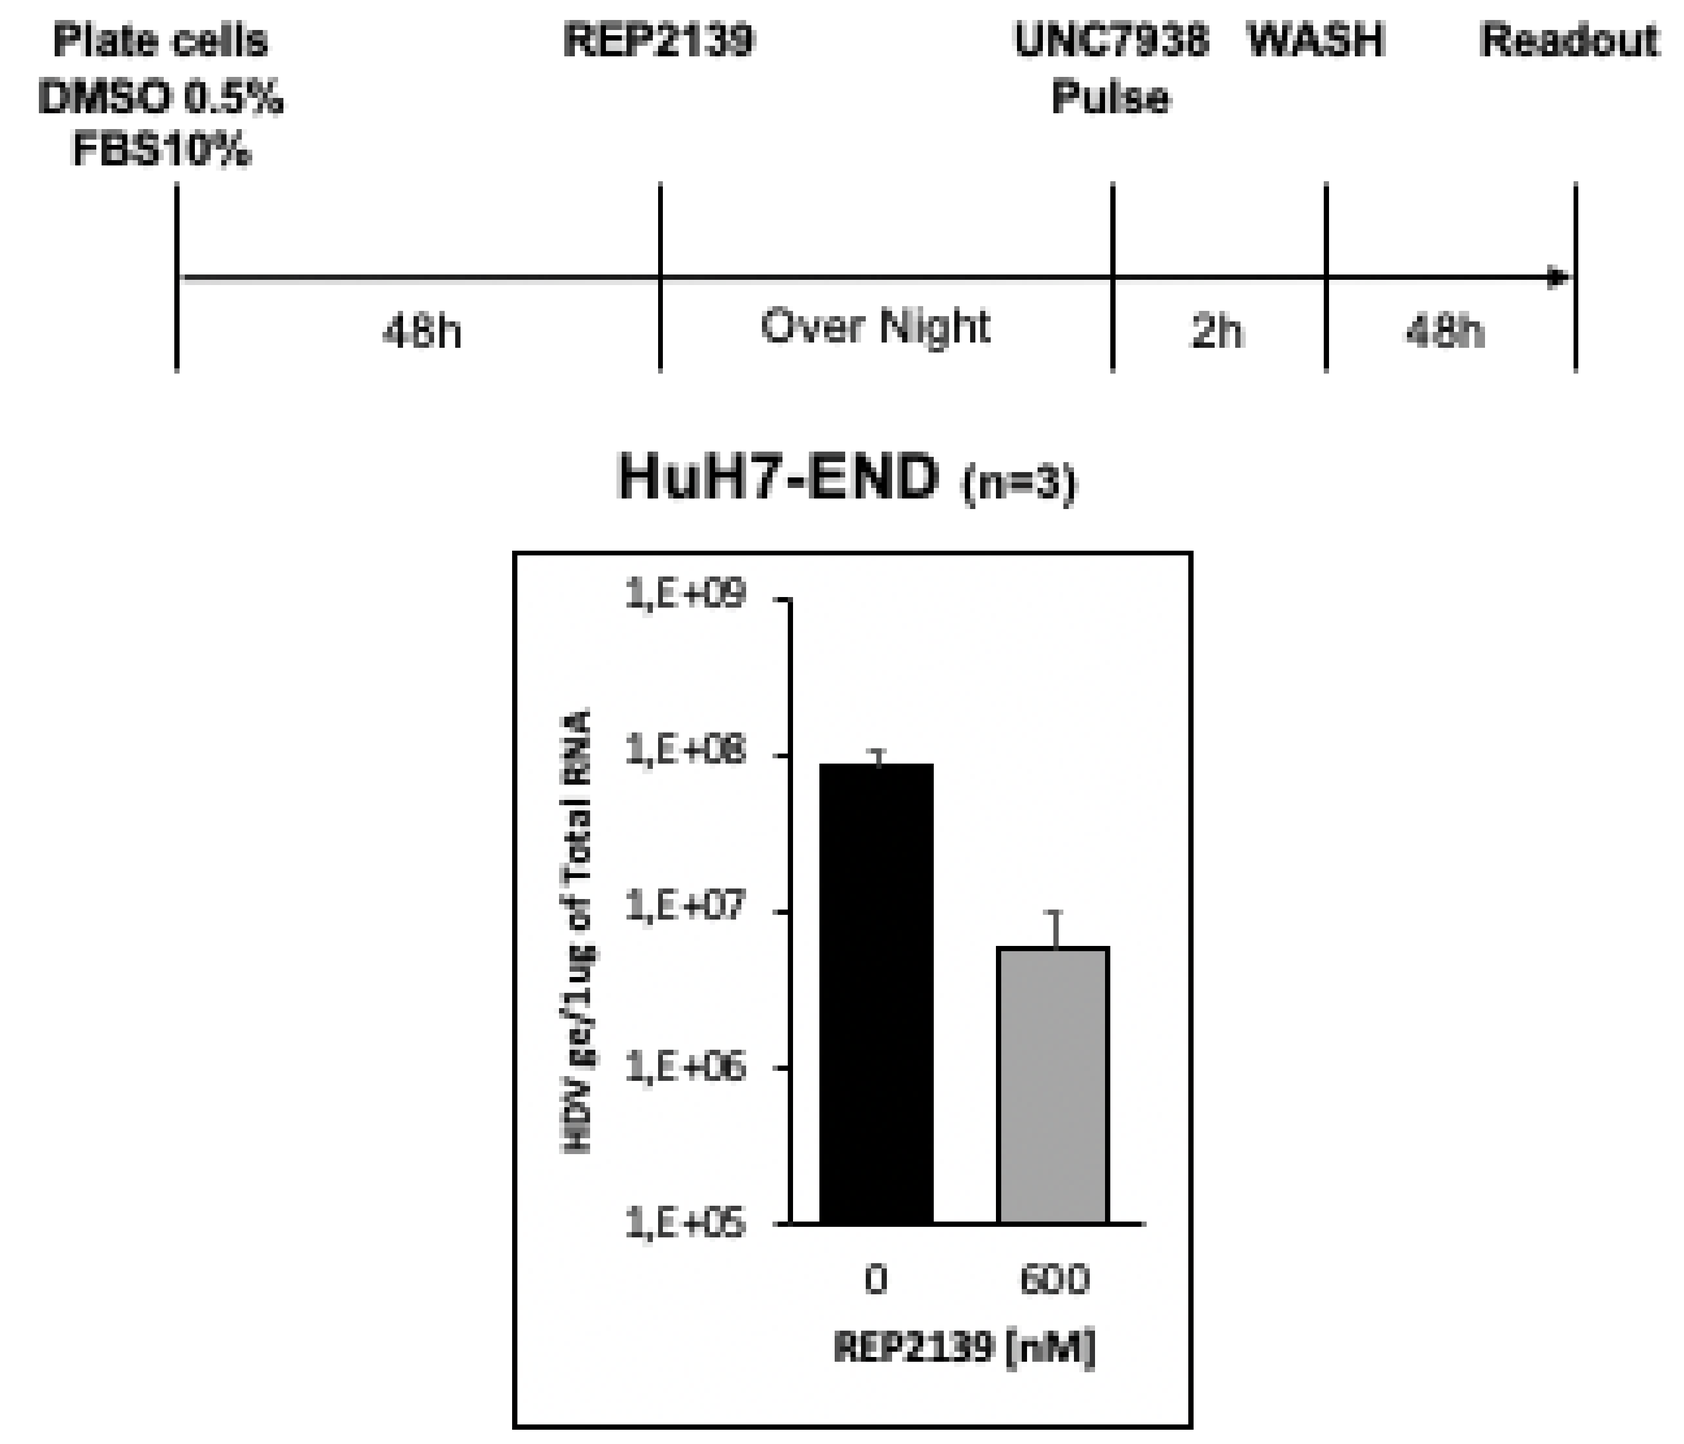
Plate cells DMSO 0.5% FBS10%
REP2139
UNC7938 Pulse
WASH
Readout
48h
Over Night
2h
48h
HuH7-END (n=3)
HDV ge/1ug of Total RNA
0 600
REP2139 [nM]

Results 2. REP2139-Mg inhibits the HDV replication in a dose-dependent manner. At higher dosage it accumulates in the endosomes and requires higher concentration of UN7938 to be released

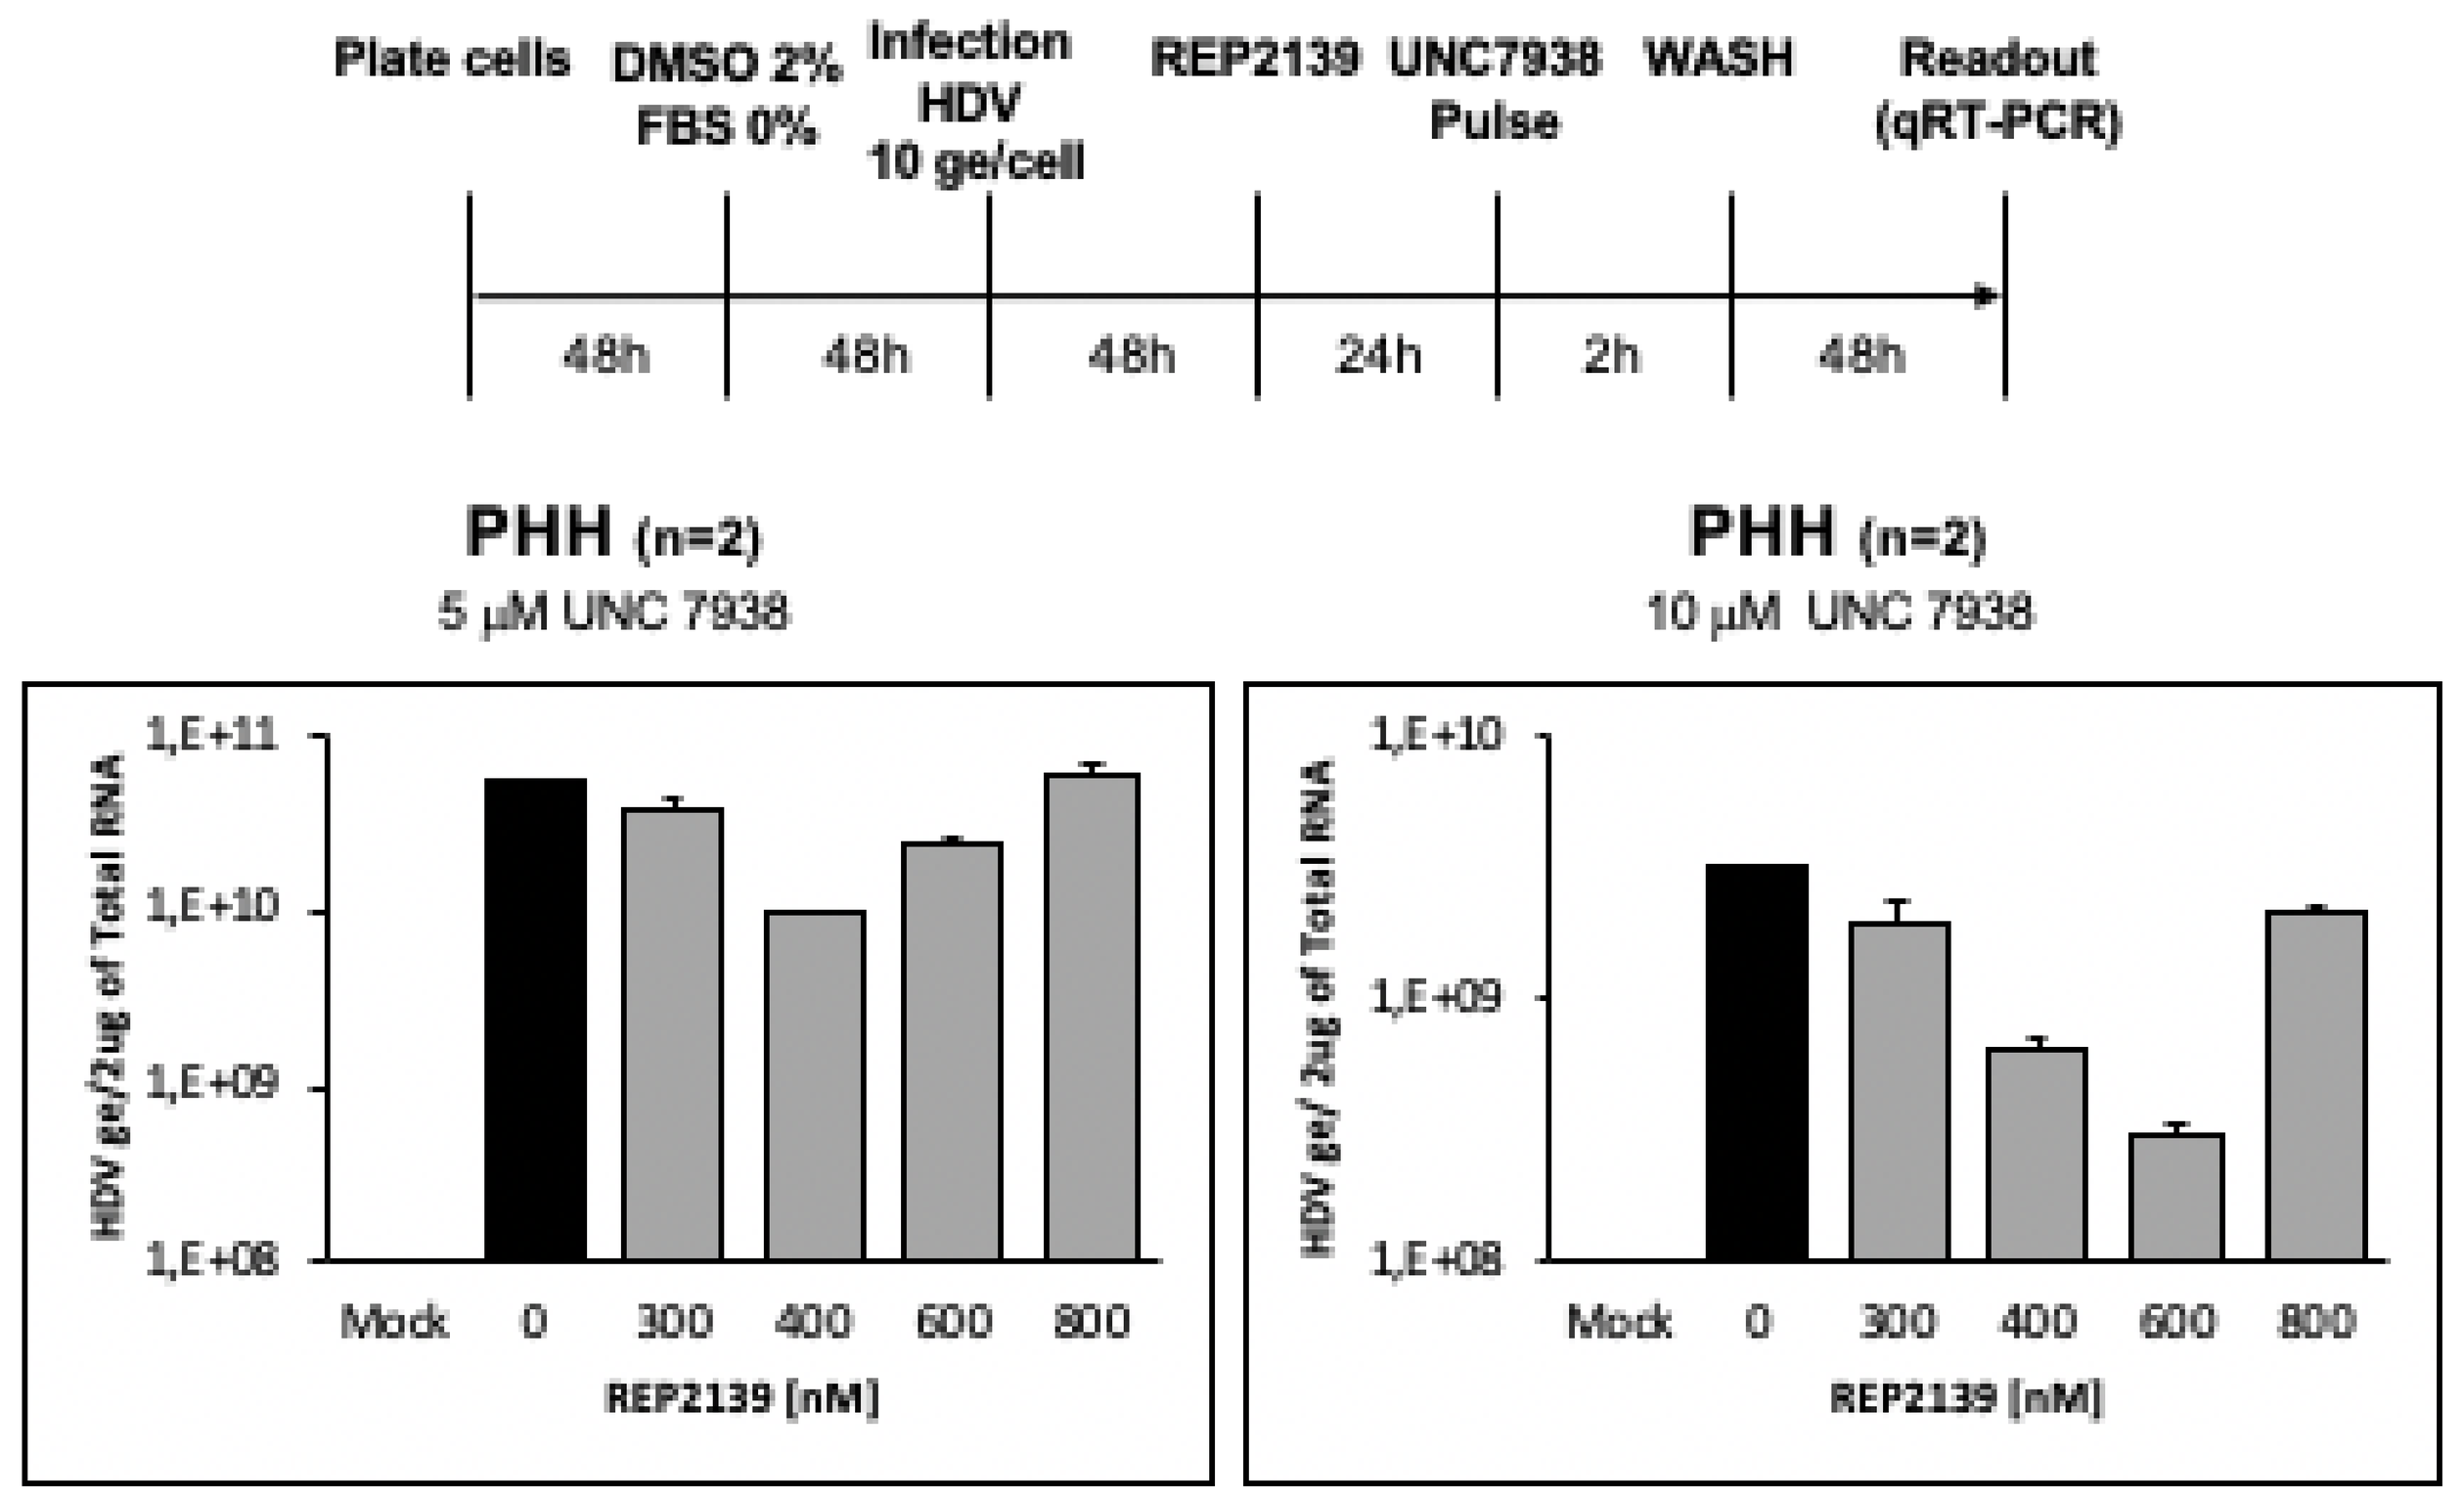
Plate cells
DMSO 2% FBS 0%
Infection HDV 10 ge/cell
REP2139
UNC7938 Pulse
WASH
Readout (qRT-PCR)
48h 48h 48h 24h 2h 48h
PHH (n=2)
5 µM UNC 7938
PHH (n=2)
10 µM UNC 7938
HDV ge/2ug of Total RNA
Mock 0 300 400 600 800
REP2139 [nM]

Results 3. REP 2139-Mg reduces both genomic (≈1Log) and antigenomic (≈1.5Log) HDV RNA

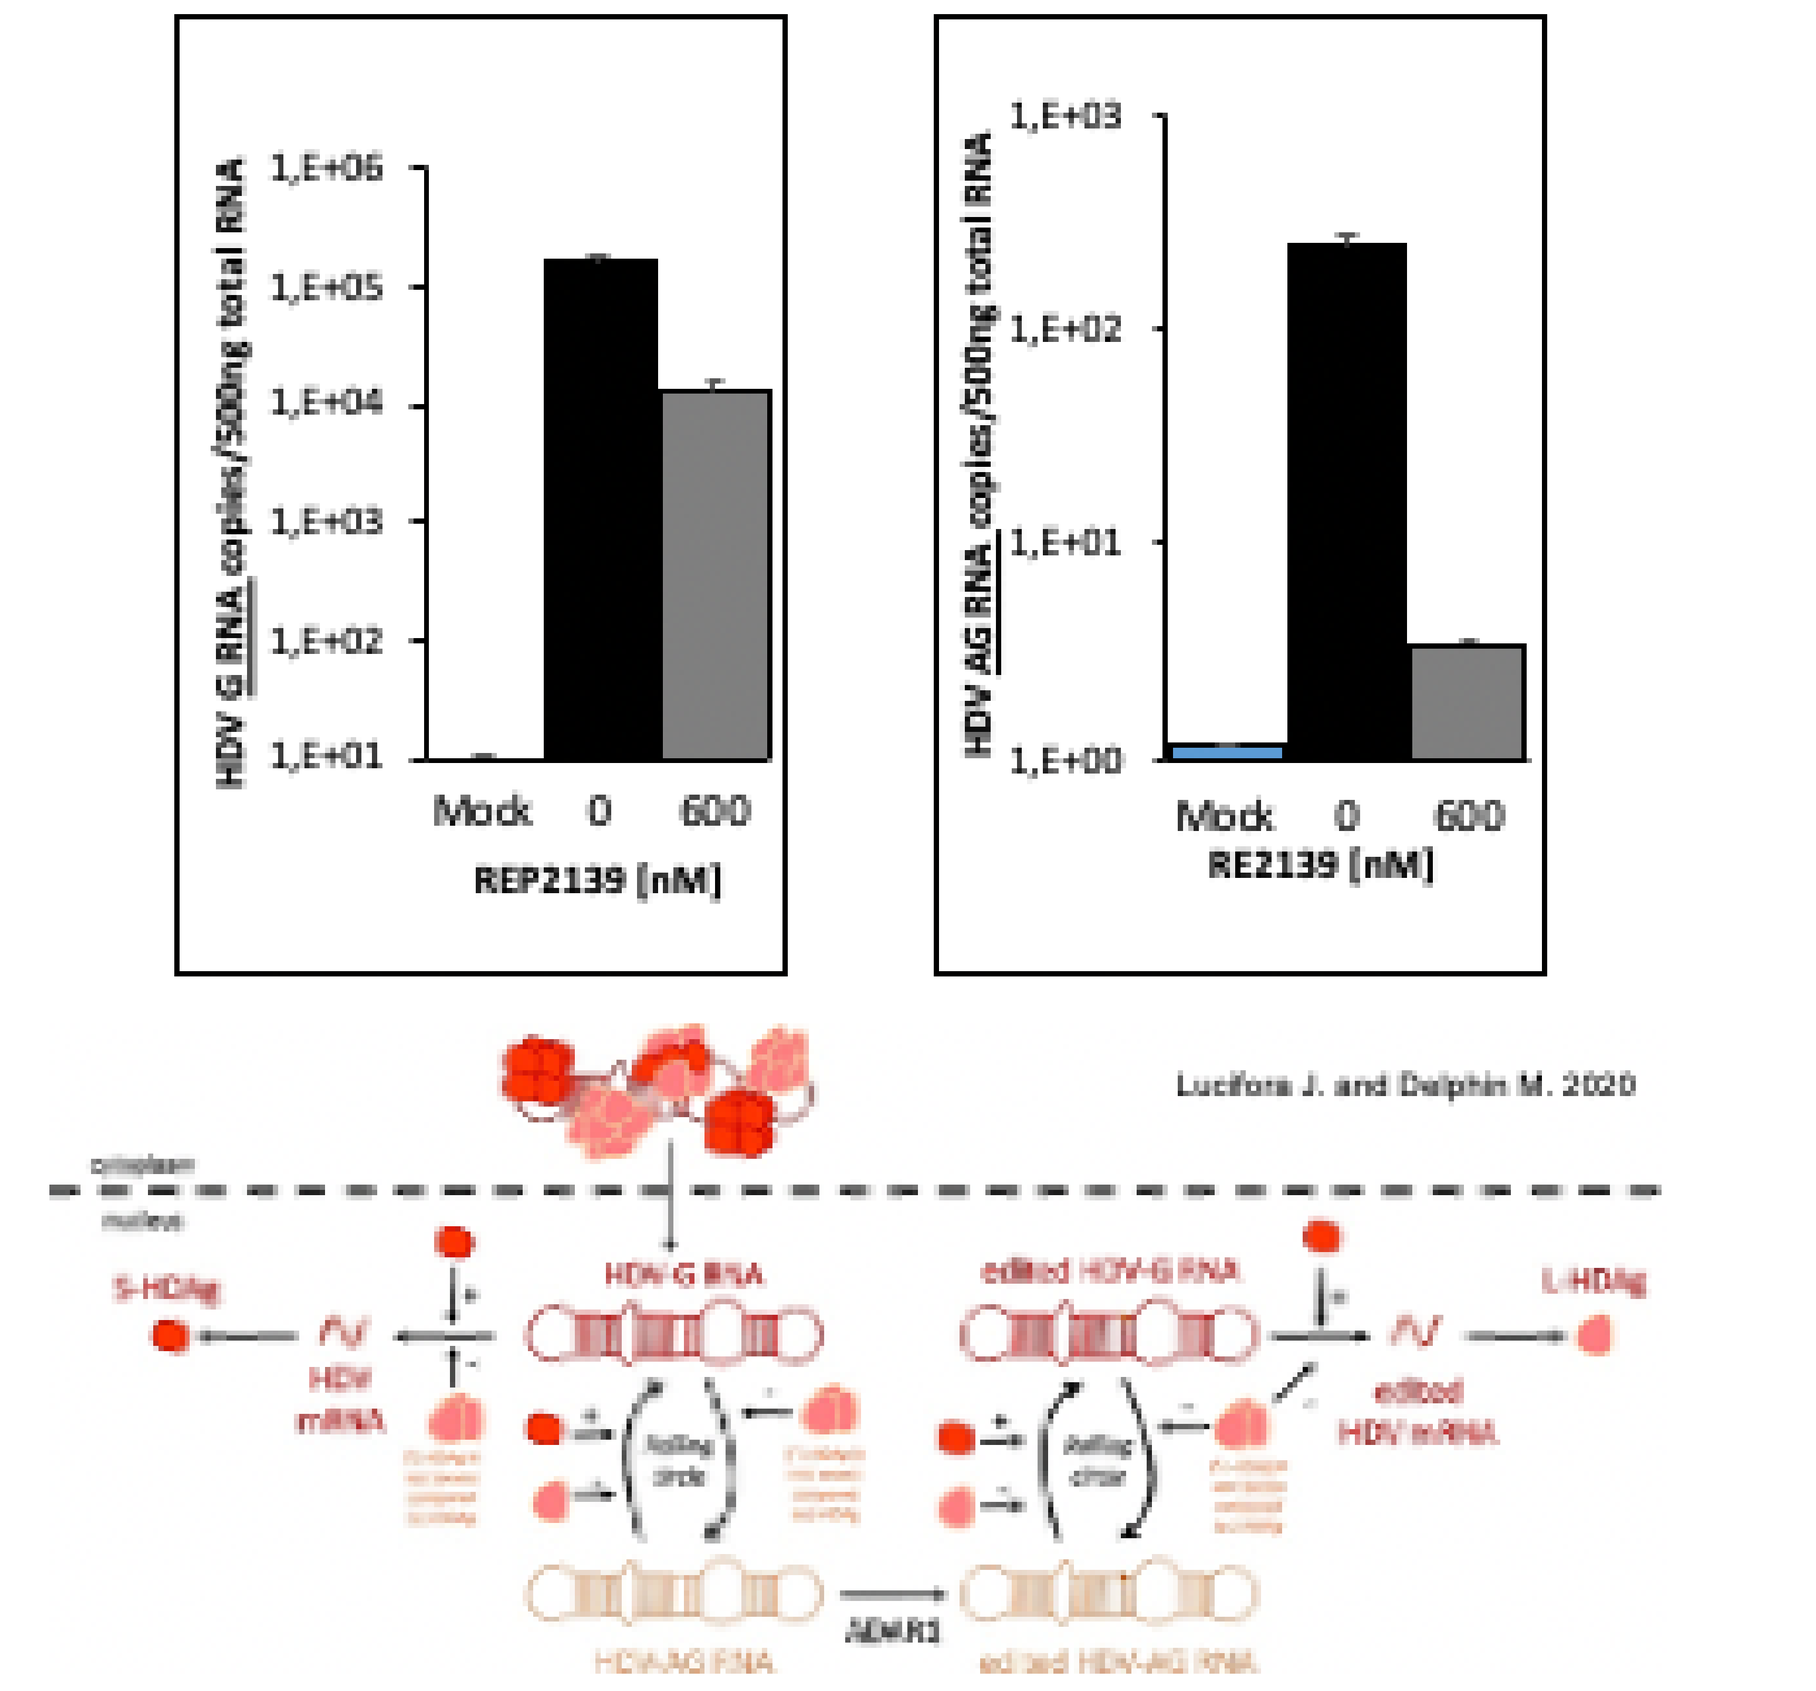
HDV G RNA copies/500ng total RNA
HDV AG RNA copies/500ng total RNA
Mock 0 600
REP2139 [nM]
RE2139 [nM]
Lucifora J. and Delphin M. 2020
S-HDAg
L-HDAg
HDV mRNA
HDV-G RNA
edited HDV-G RNA
edited HDV mRNA
HDV-AG RNA
ADAR1
edited HDV-AG RNA

Results 4. A single REP 2139-Mg dose also reduced the intranuclear association of HDV RNA with HDAg by ~60% in HepG2-NTCP and by 80% in PHH, **without changing the HDAg protein levels**.

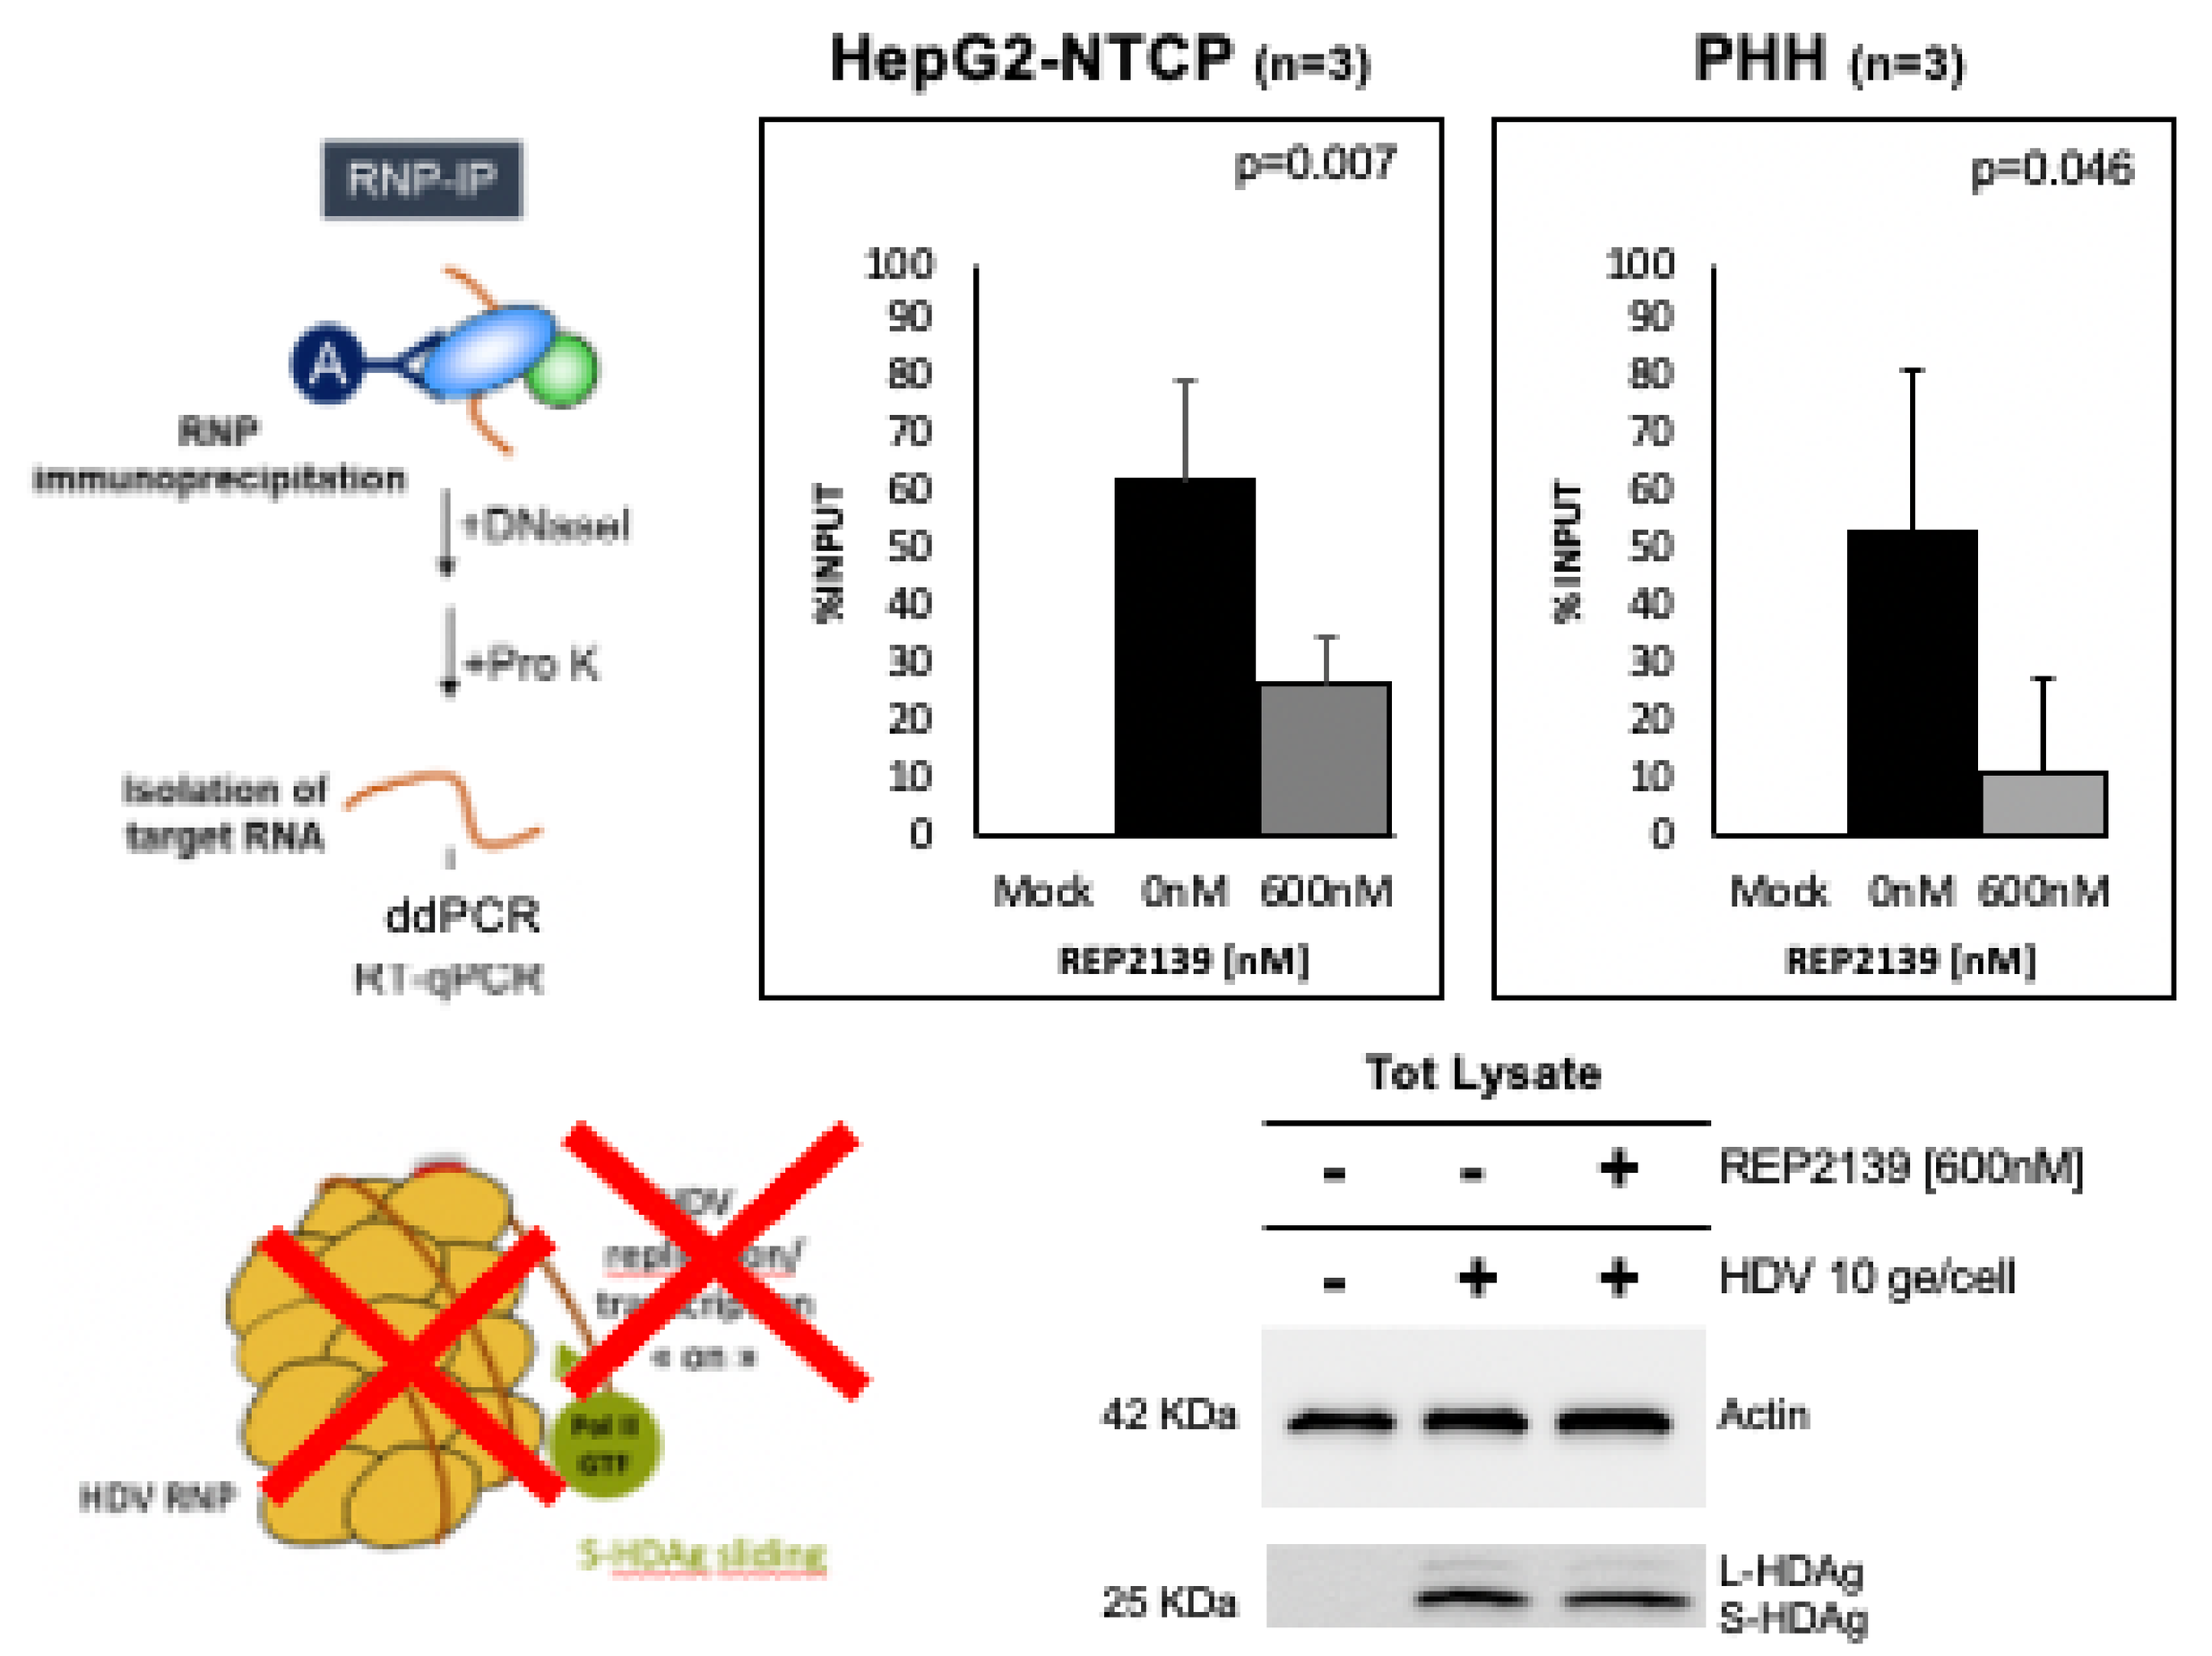
RNP-IP
RNP immunoprecipitation
+DNaseI
+Pro K
Isolation of target RNA
ddPCR
RT-qPCR
HDV RNP
S-HDAg sliding
HepG2-NTCP (n=3)
p=0.007
PHH (n=3)
p=0.046
%INPUT
Mock 0nM 600nM
REP2139 [nM]
Tot Lysate
REP2139 [600nM]
HDV 10 ge/cell
42 KDa
Actin
25 KDa
L-HDAg
S-HDAg

Results 5. REP 2139-Mg inhibits HDV replication in HBV-HDV coinfected PHHs. We confirm in HBV-HDV coinfected PHHs that REP 2139 inhibits HBsAg secretion without affecting intracellular pgRNA levels or HBeAg secretion

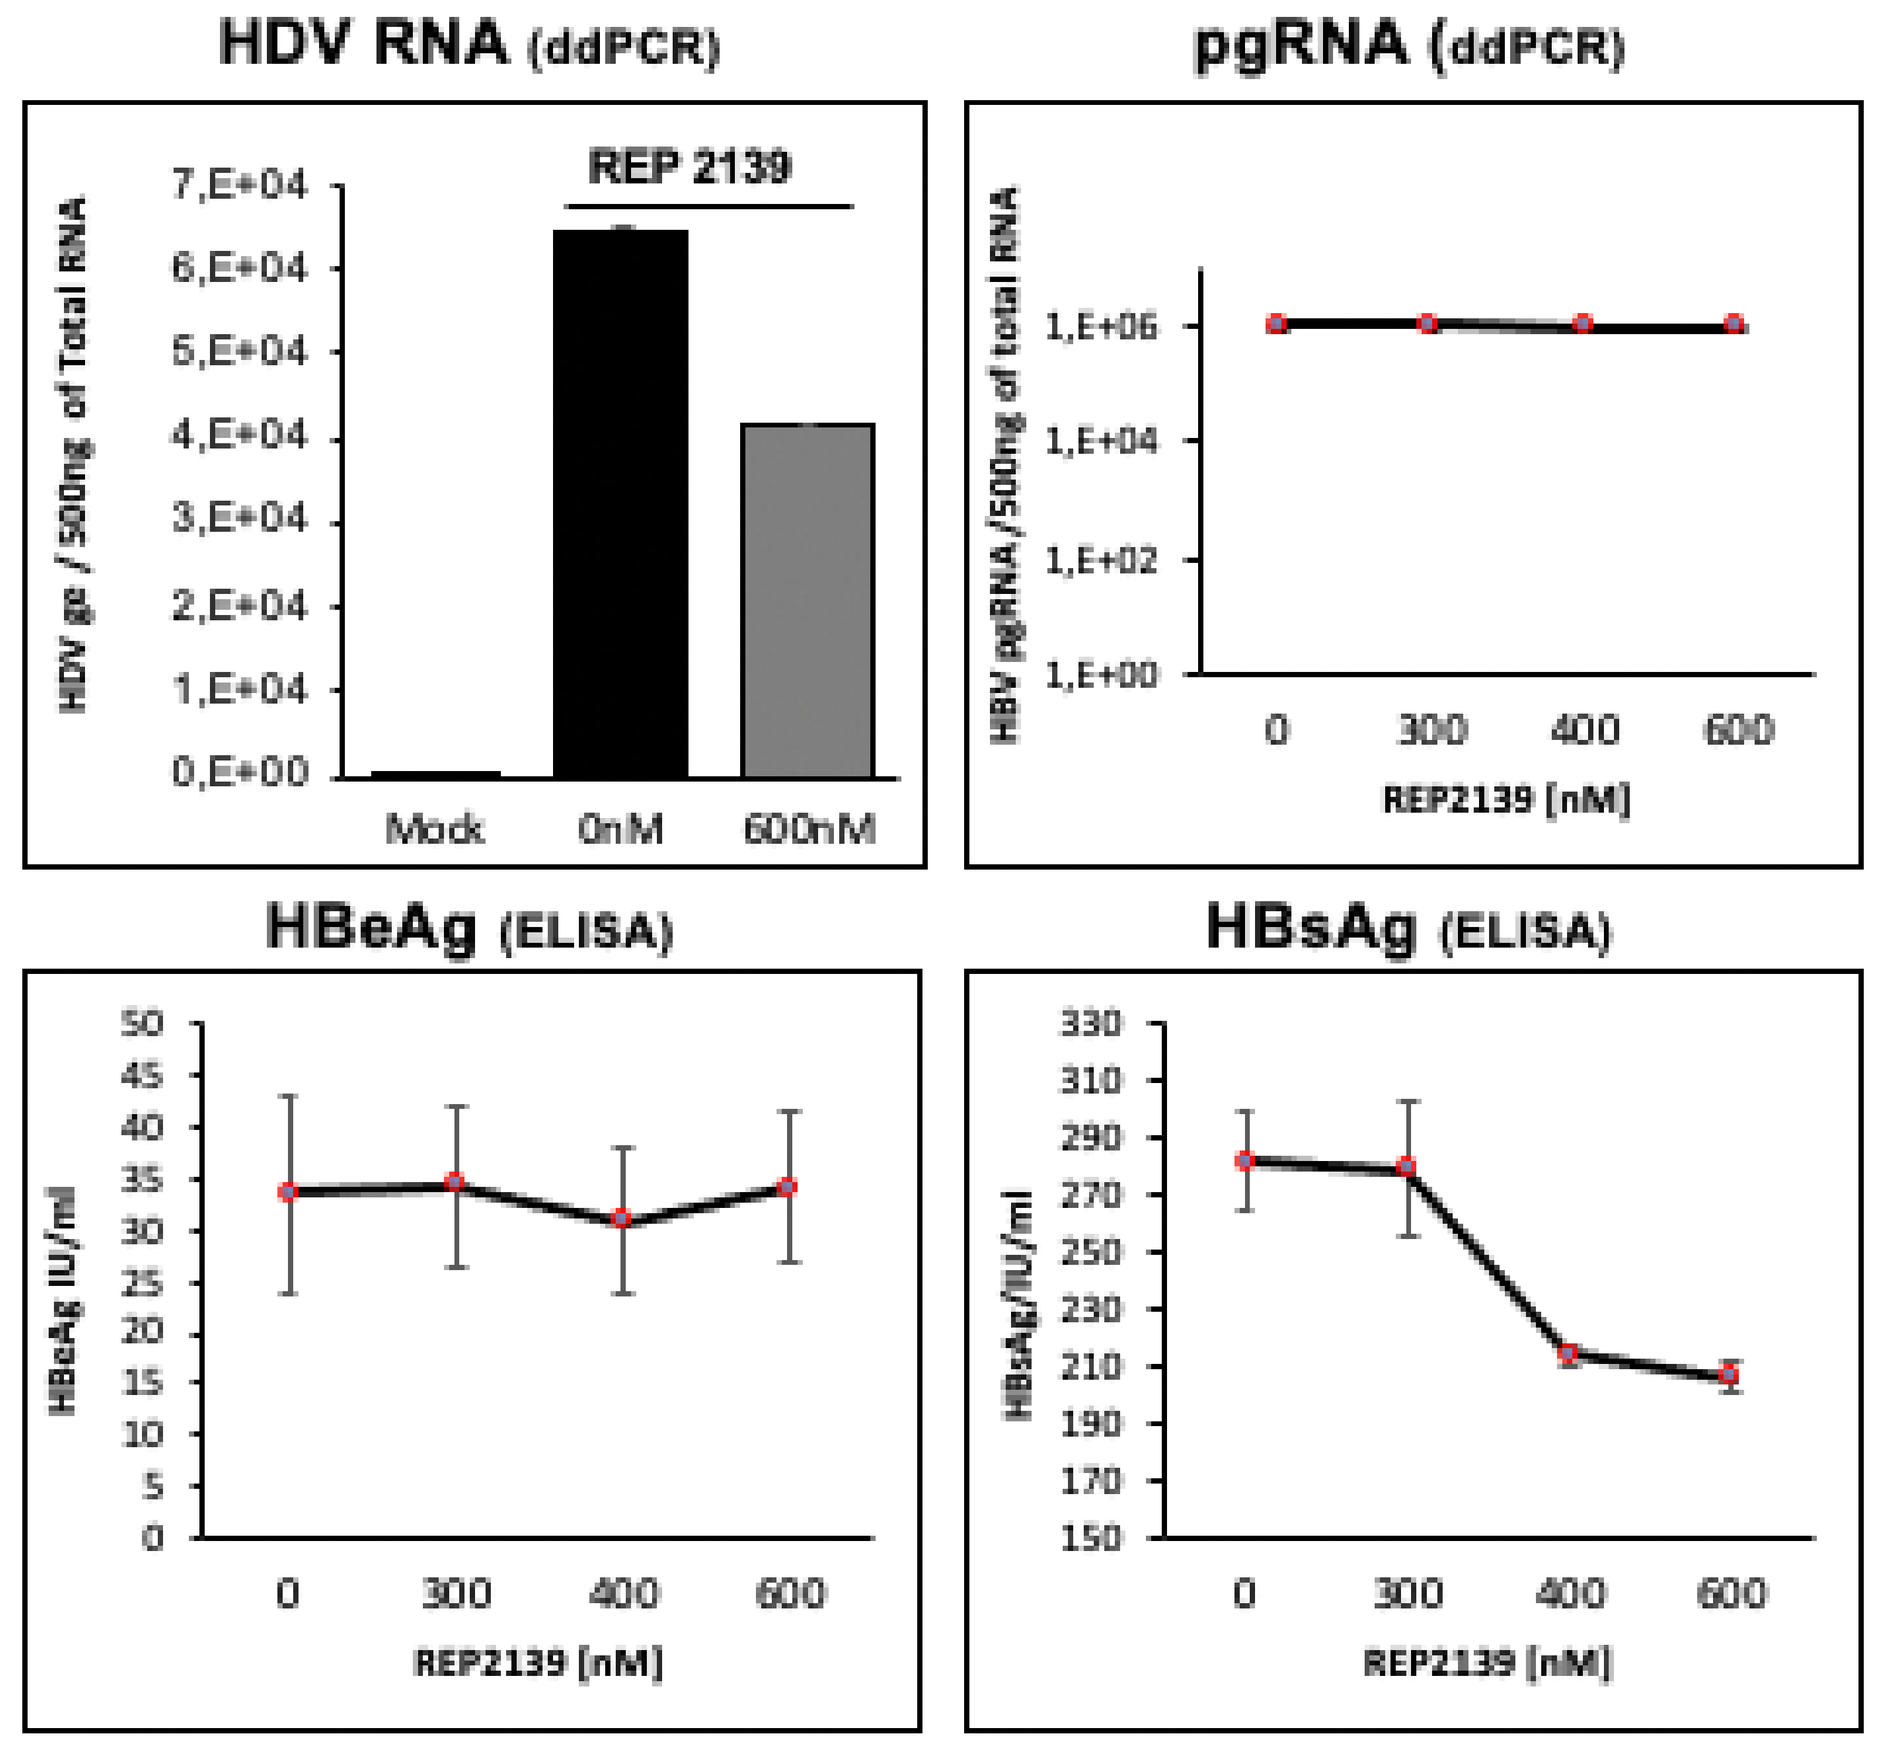
HDV RNA (ddPCR)
pgRNA (ddPCR)
REP 2139
HDV ge / 500ng of Total RNA
HBV pgRNA /500ng of total RNA
Mock 0nM 600nM
0 300 400 600
REP2139 [nM]
HBeAg (ELISA)
HBsAg (ELISA)
HBeAg IU/ml
HBsAg/IU/ml