Pangenomic antiviral effect of REP 2139 in CRISPR/Cas9 engineered cell lines expressing hepatitis B surface antigen.

AASLD Nov. 10-14, 2023 The Liver Meeting®

Abstract # 1479-C

Lena Angelo[1], Matthieu Blanchet[1,2], Andrew Vaillant[2], Patrick Labonté[1]

1.Institut National de la Recherche Scientifique – Centre Armand Frappier Santé Biotechnologie, Laval, Canada.
2.Replicor Inc. Montréal, Canada.

ABSTRACT

Hepatitis B virus remains a global health problem with 296 million people living with chronic infection who are at increased risk of developing liver inflammation, cirrhosis and hepatocellular carcinoma. Subviral particles (SVP) are produced in large excess over Dane particles in patients and are the major source of HBsAg. SVPs inhibit /exhaust the immune response to HBV infection. Functional cure which can occur after the reestablishment of the immune control of HBV requires the clearance of HBsAg from blood of patient. Nucleic acid polymers (NAPs) inhibit the assembly/release of SVP, resulting in rapid clearance of HBsAg from circulation in vitro and in human studies. However, their efficacy has only been demonstrated in limited genotypes in phase IIA clinical trials. HBV exists as nine main genotypes (A to I), and our study assesses the activity of the lead NAP (REP 2139) in the most prevalent genotypes (A, B, C, D, E, G), which account for over 96% of chronic HBV infection.

HBsAg ORFs from the various genotypes were inserted into the AAVS1 safe-harbor of HepG2 cells using CRISPR/Cas9 knock-in. A cell line producing the D144A vaccine escape mutant was also engineered. The secretion of HBsAg into these new genotype cell lines (GCLs) was confirmed by immunofluorescence and ELISA. The antiviral activity of REP 2139 was then assessed in these GCLs by ELISA.

The results show an efficient inhibition of HBsAg secretion in all GCLs.

We demonstrate that REP 2139 exerts an antiviral effect in all genotypes and serotypes tested in this study, including in the vaccine escape mutant D144A. EC50 for every genotype assessed in this study are in the nanomolar range, which is in line with previous in vitro and clinical studies.

Our results suggest that REP 2139 has a pangenomic antiviral effect, as well as an antiviral effect on escape mutants.

METHODS

pAAVS1-puro-HBsAg-HBx

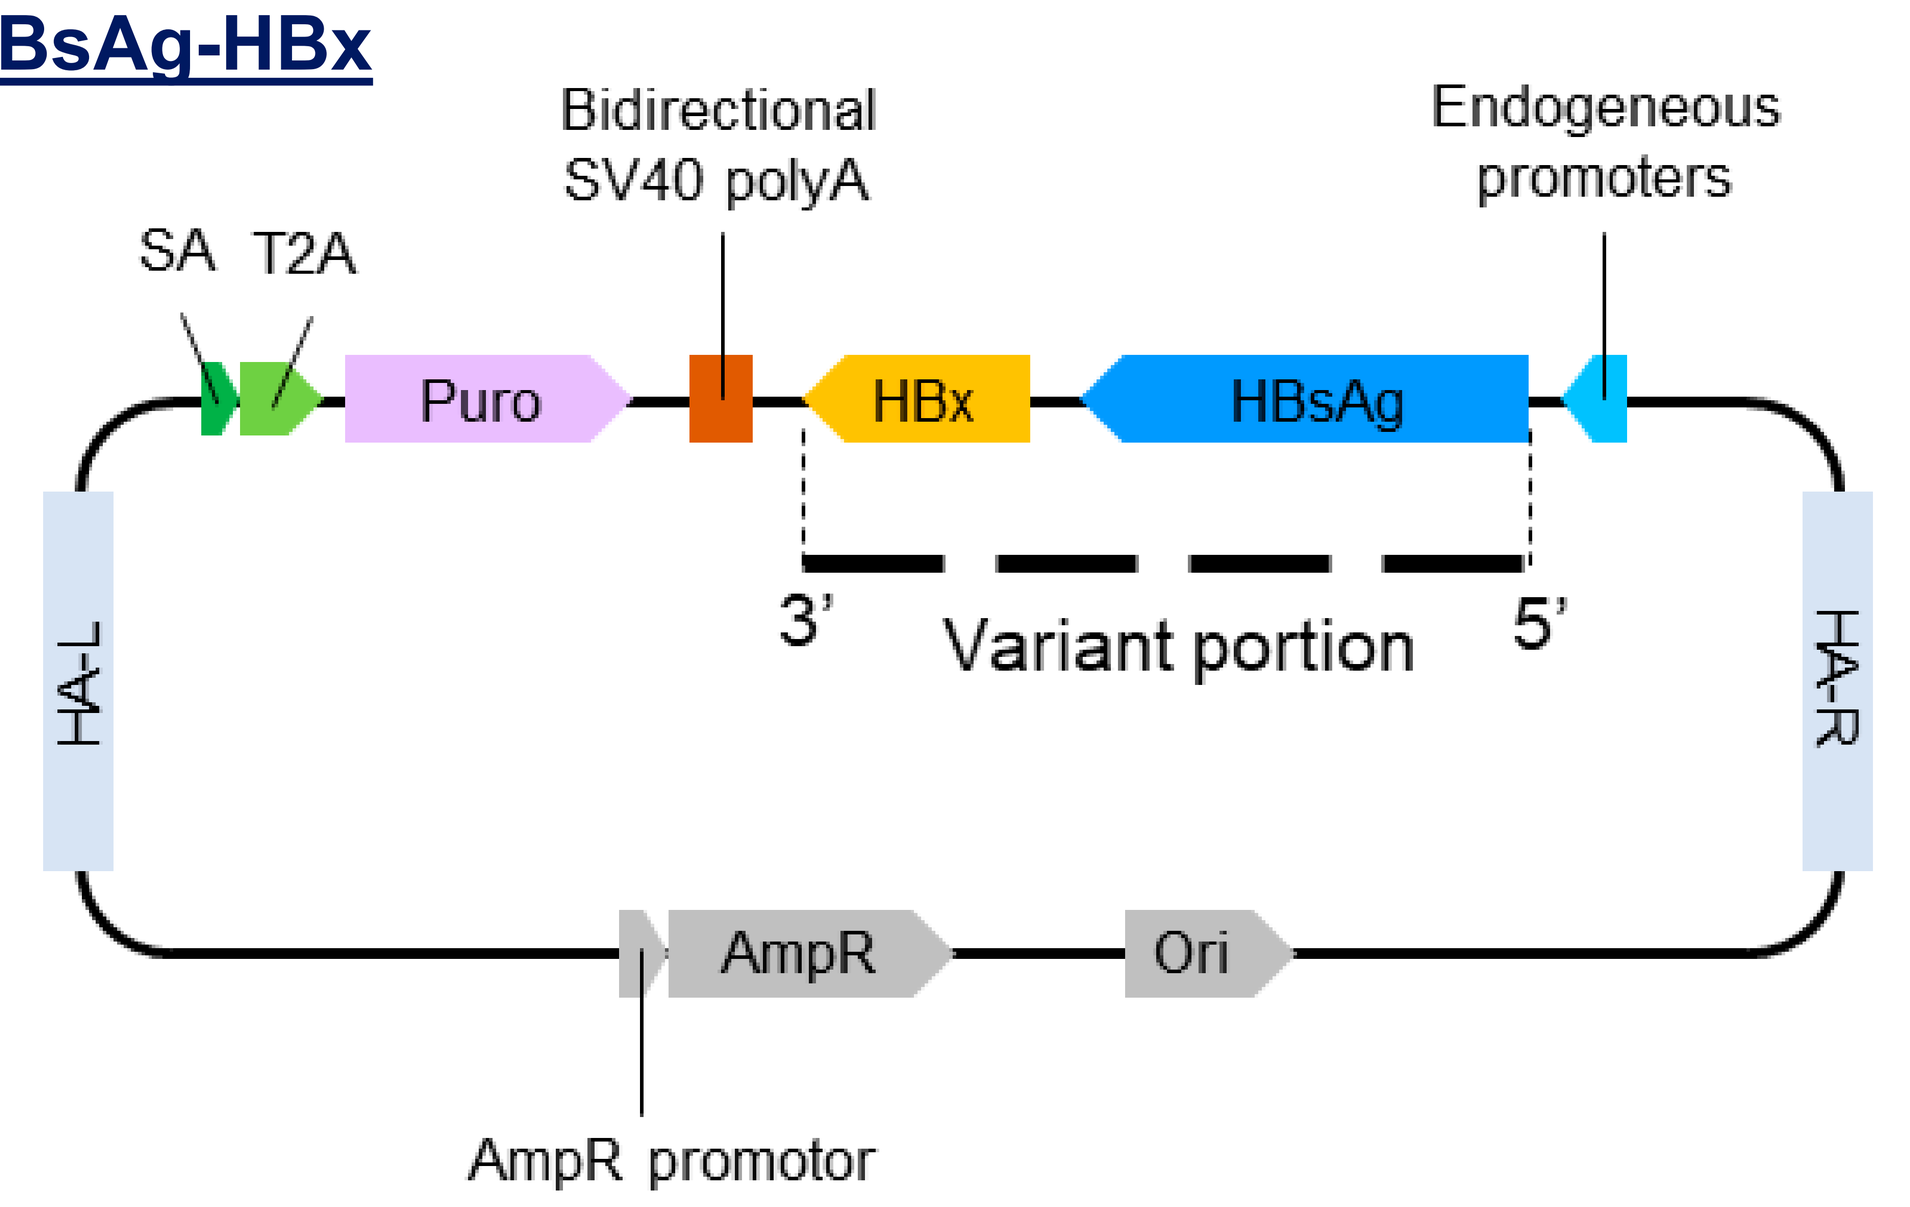

Bidirectional SV40 polyA
Endogeneous promoters
SA
T2A
Puro
HBx
HBsAg
3'
Variant portion
5'
HA-L
HA-R
AmpR
Ori
AmpR promotor

Donor plasmid construction used to create Genotype Cell Lines (GCLs):
Donor plasmids were created from the plasmid AAVS1_Puro_Tet3G_3xFLAG_Twin_Strep (Addgene #92099) cleaved with SalI and NsiI with the subsequent cleaved fragment replaced with the HBsAg ORF +/- the HBx ORF from pT7HB2.7 plasmids (pT7HB2.7 plasmids containing PreS1, PreS2 and S genes from various genotypes and the X gene from genotype D ayw3).
These plasmids were used as donors for the CRISPR/Cas9 knock-in to generate the GCLs.

CRISPR/Cas9 knock-in:
For each genotype, 5×10^5 HepG2 cells/well were seeded on collagen-coated 6-well plates and transfected the next day with Cas9/sgRNA coding plasmid and pAAVS1-puro-HBsAg-HBx (for each genotype) at a ratio of 1:1, using Lipofectamine™ 3000. Cells were cultured for 14 days before addition of puromycin (0.5 µL/mL). When transfected cells were confluent, cells were trypsinized, plated at various densities, and cultured with 0.5 µL/mL of puromycin for 52 days. Remaining living cells were trypsinized, pooled together as population cell lines, and HBsAg secretion in these cell lines was assessed before performing further experiments.

HBsAg detection:
Quantification of HBsAg from supernatant of GCLs was conducted using the GS HBsAg EIA 3.0 Kit. Confocal fluorescence microscopy was performed to observe intracellular expression of HBsAg.

RESULTS

A

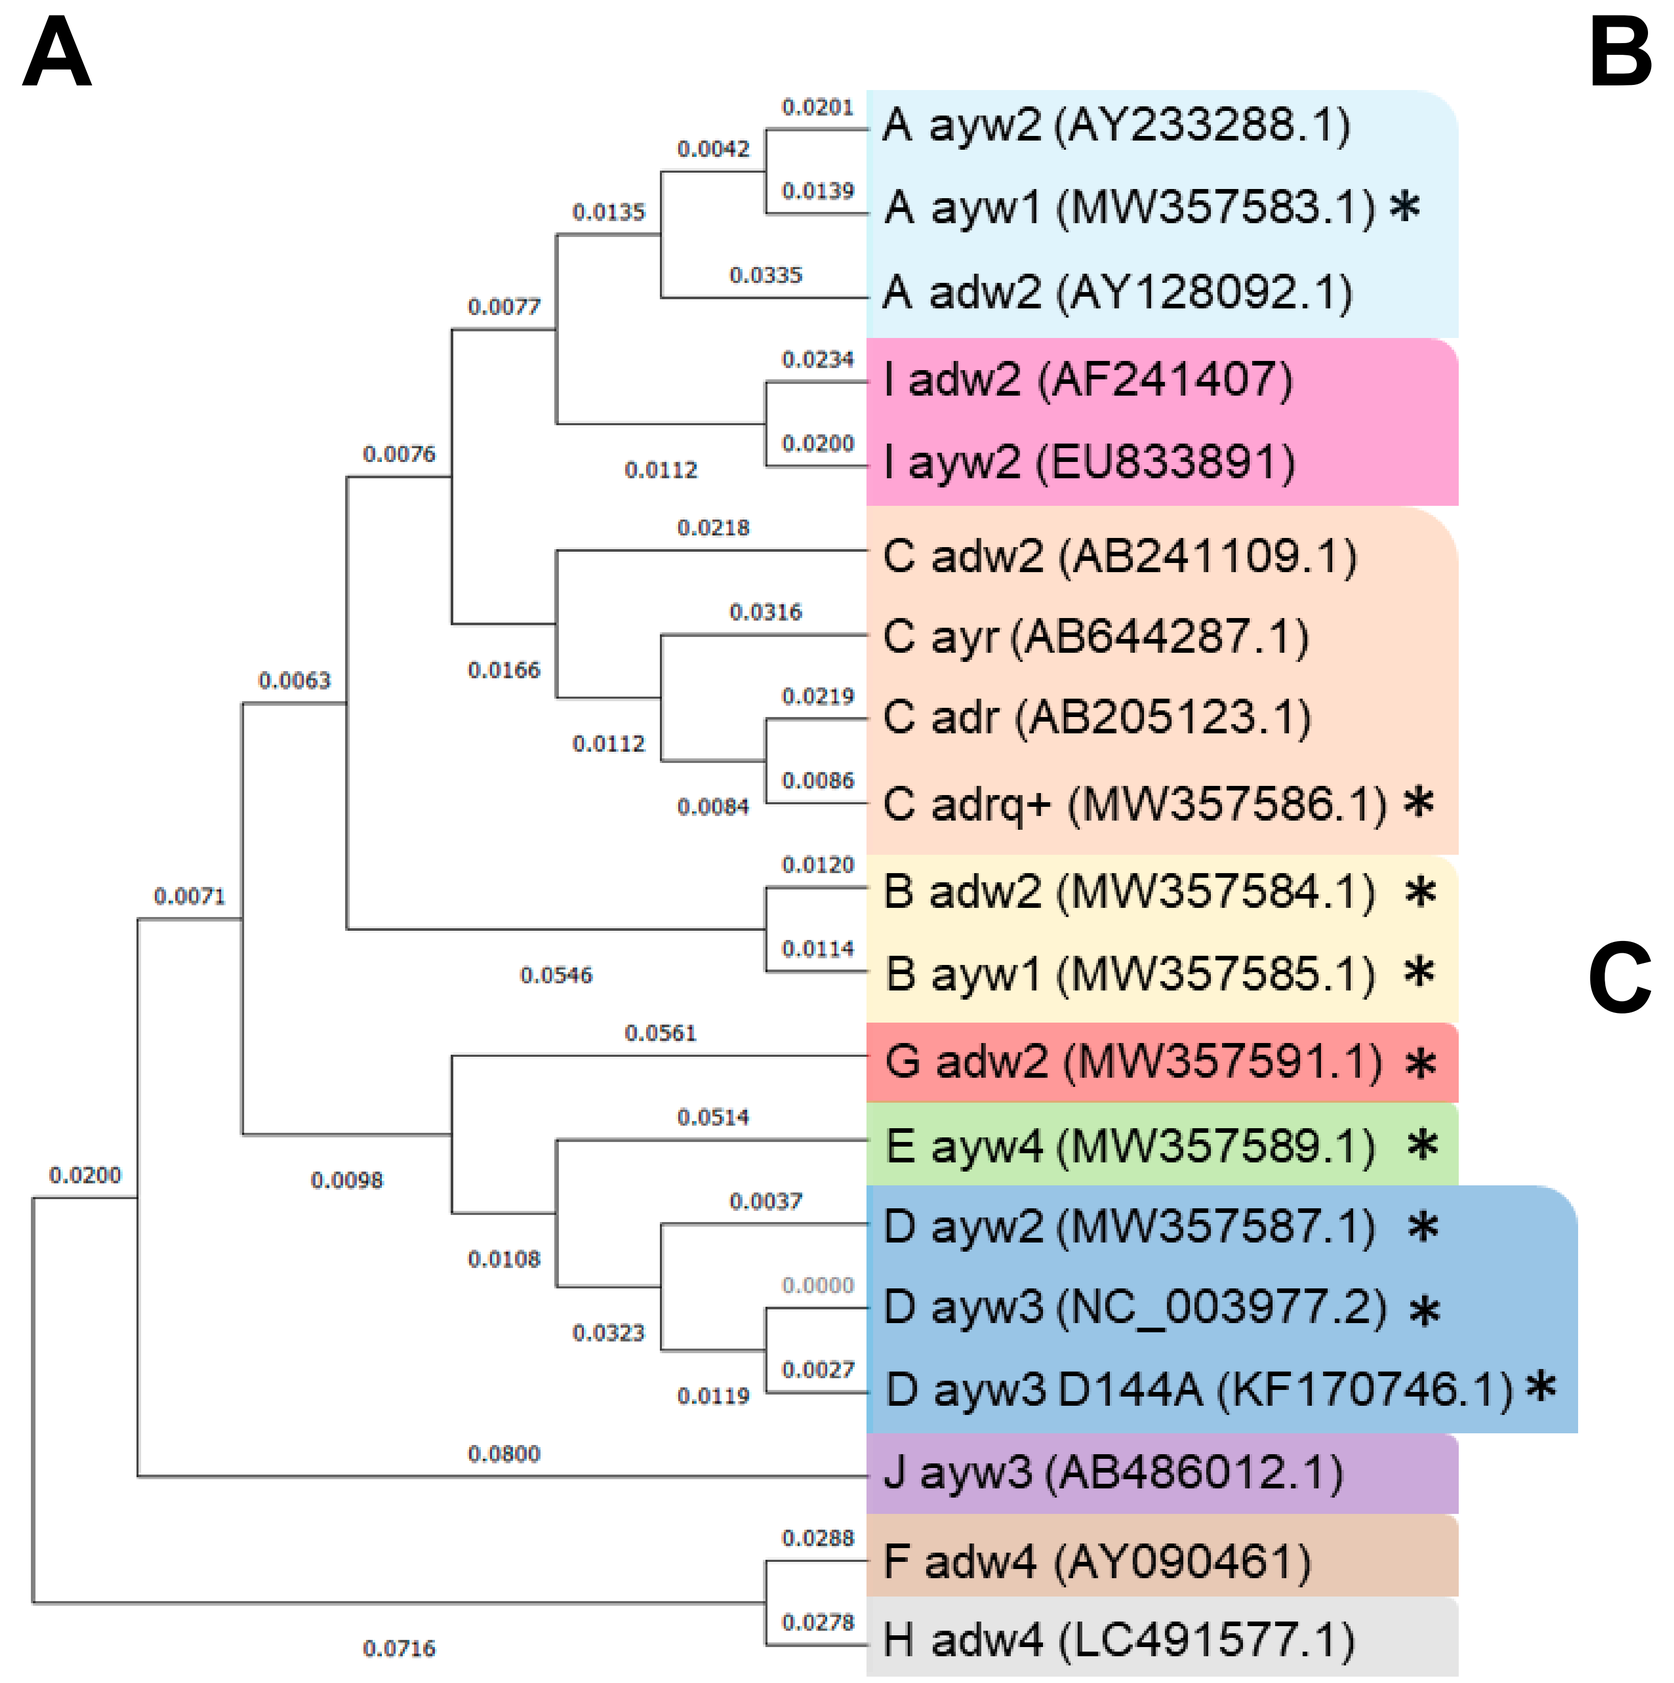

A ayw2 (AY233288.1)
A ayw1 (MW357583.1) *
A adw2 (AY128092.1)
I adw2 (AF241407)
I ayw2 (EU833891)
C adw2 (AB241109.1)
C ayr (AB644287.1)
C adr (AB205123.1)
C adrq+ (MW357586.1) *
B adw2 (MW357584.1) *
B ayw1 (MW357585.1) *
G adw2 (MW357591.1) *
E ayw4 (MW357589.1) *
D ayw2 (MW357587.1) *
D ayw3 (NC_003977.2) *
D ayw3 D144A (KF170746.1) *
J ayw3 (AB486012.1)
F adw4 (AY090461)
H adw4 (LC491577.1)

B

Existing genotypes	
A	✓
B	✓
C	✓
D	✓
E	✓
F	
G	✓
H	
I	

C

Existing serotypes	
adw2	✓
adw3	
adw4	
ayw1	✓
ayw2	✓
ayw3	✓
ayw4	✓
adr	✓
ayr	

Fig 1. Phylogenic classification of the L-HBsAg protein ORF according to genotypes and serotypes.
(A) Phylogenic tree for one of each genotype/serotype couple existing for every genotype, (*) marks those evaluated in this study.. (B) Existing HBV genotypes identified, (✓) marks those included in this study. (C) Existing HBV serotypes identified, (✓) marks those studied in this study.

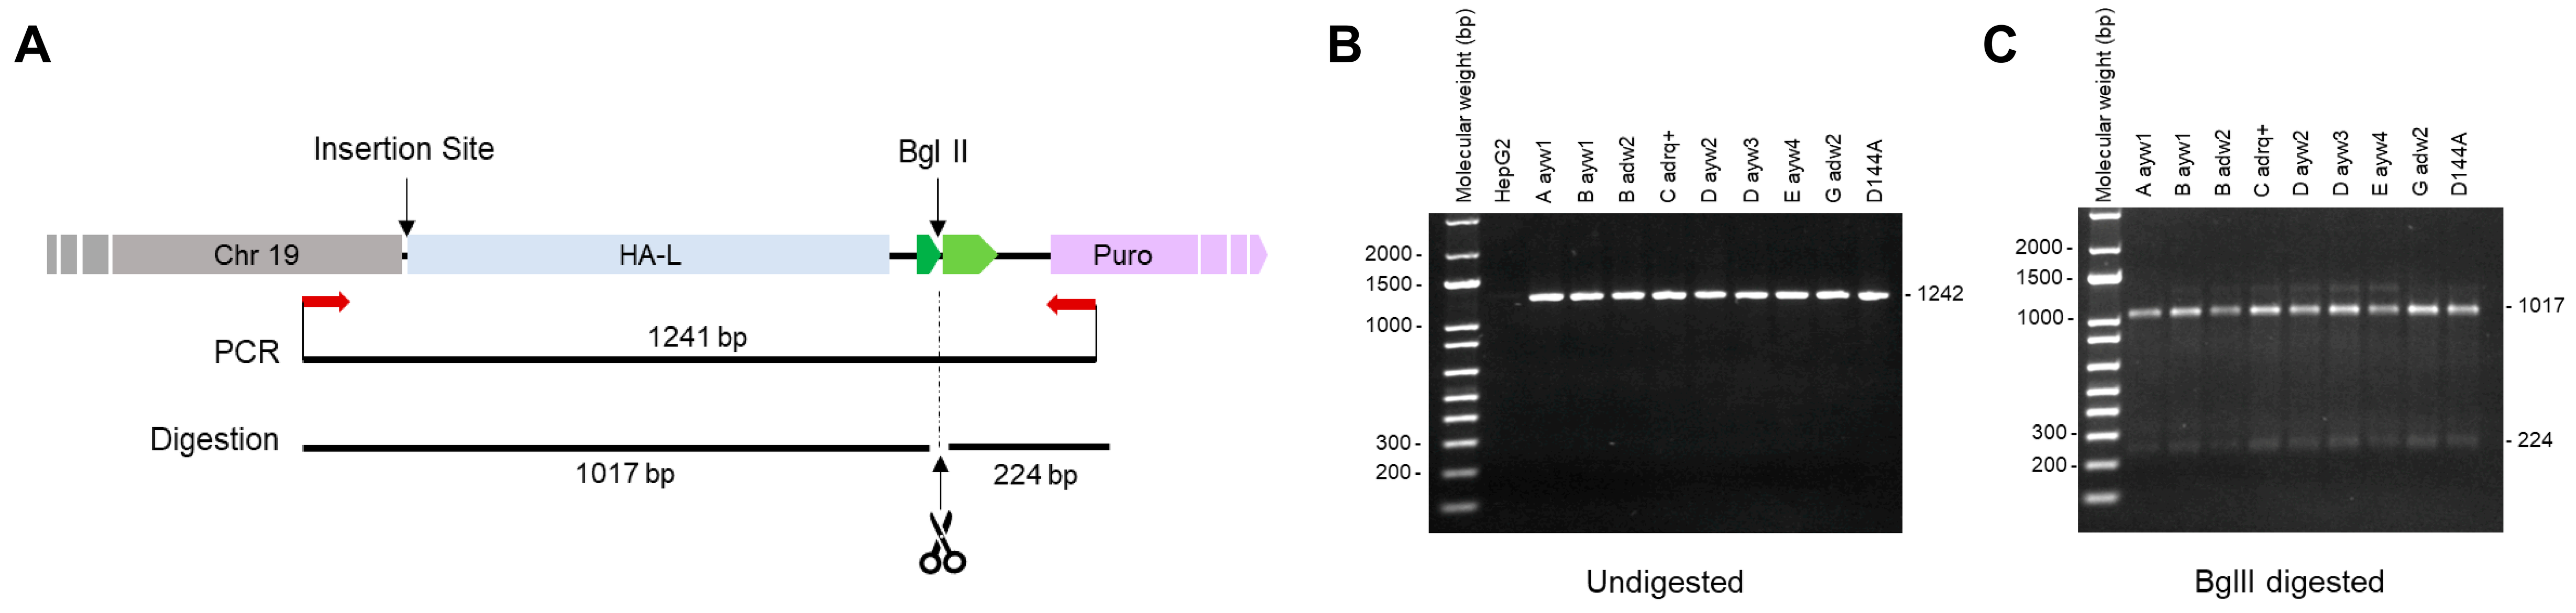

A
Insertion Site
Bgl II
Chr 19
HA-L
Puro
PCR
1241 bp
Digestion
1017 bp
224 bp
B
Molecular weight (bp)
HepG2
A ayw1
B ayw1
B adw2
C adrq+
D ayw2
D ayw3
E ayw4
G adw2
D144A
2000
1500
1000
300
200
1242
Undigested
C
1017
224
BglII digested

Fig 2. Integration of HBsAg-HBx gene into the AAVS1 safe-harbor.
(A) Chromosome 19 modified by CRISPR-Cas9 to the insertion site by insert of the transgene containing the left homology-arm (HA-L) and the puromycin resistance gene is represented. Experimental design on this gDNA is as shown, primers attachment sites are indicated in red arrows, and the restriction site of BglII is also indicated. (B) Agarose gel presenting the PCR amplification product of the insertion site. (C) Agarose gel presenting the BglII digested PCR product.

Fig 3. Analysis of HBsAg expression and secretion from the indicated cell lines.
(A) Intracellular HBsAg expression analysis was conducted by confocal microscopy. Nucleus are stained in blue and HBsAg in green. (B) Secreted HBsAg from each cell line was assessed by ELISA, normalized to cell viability and to HepG2.2.15 secretion levels. Sup, supernatant; RU, relative unit.

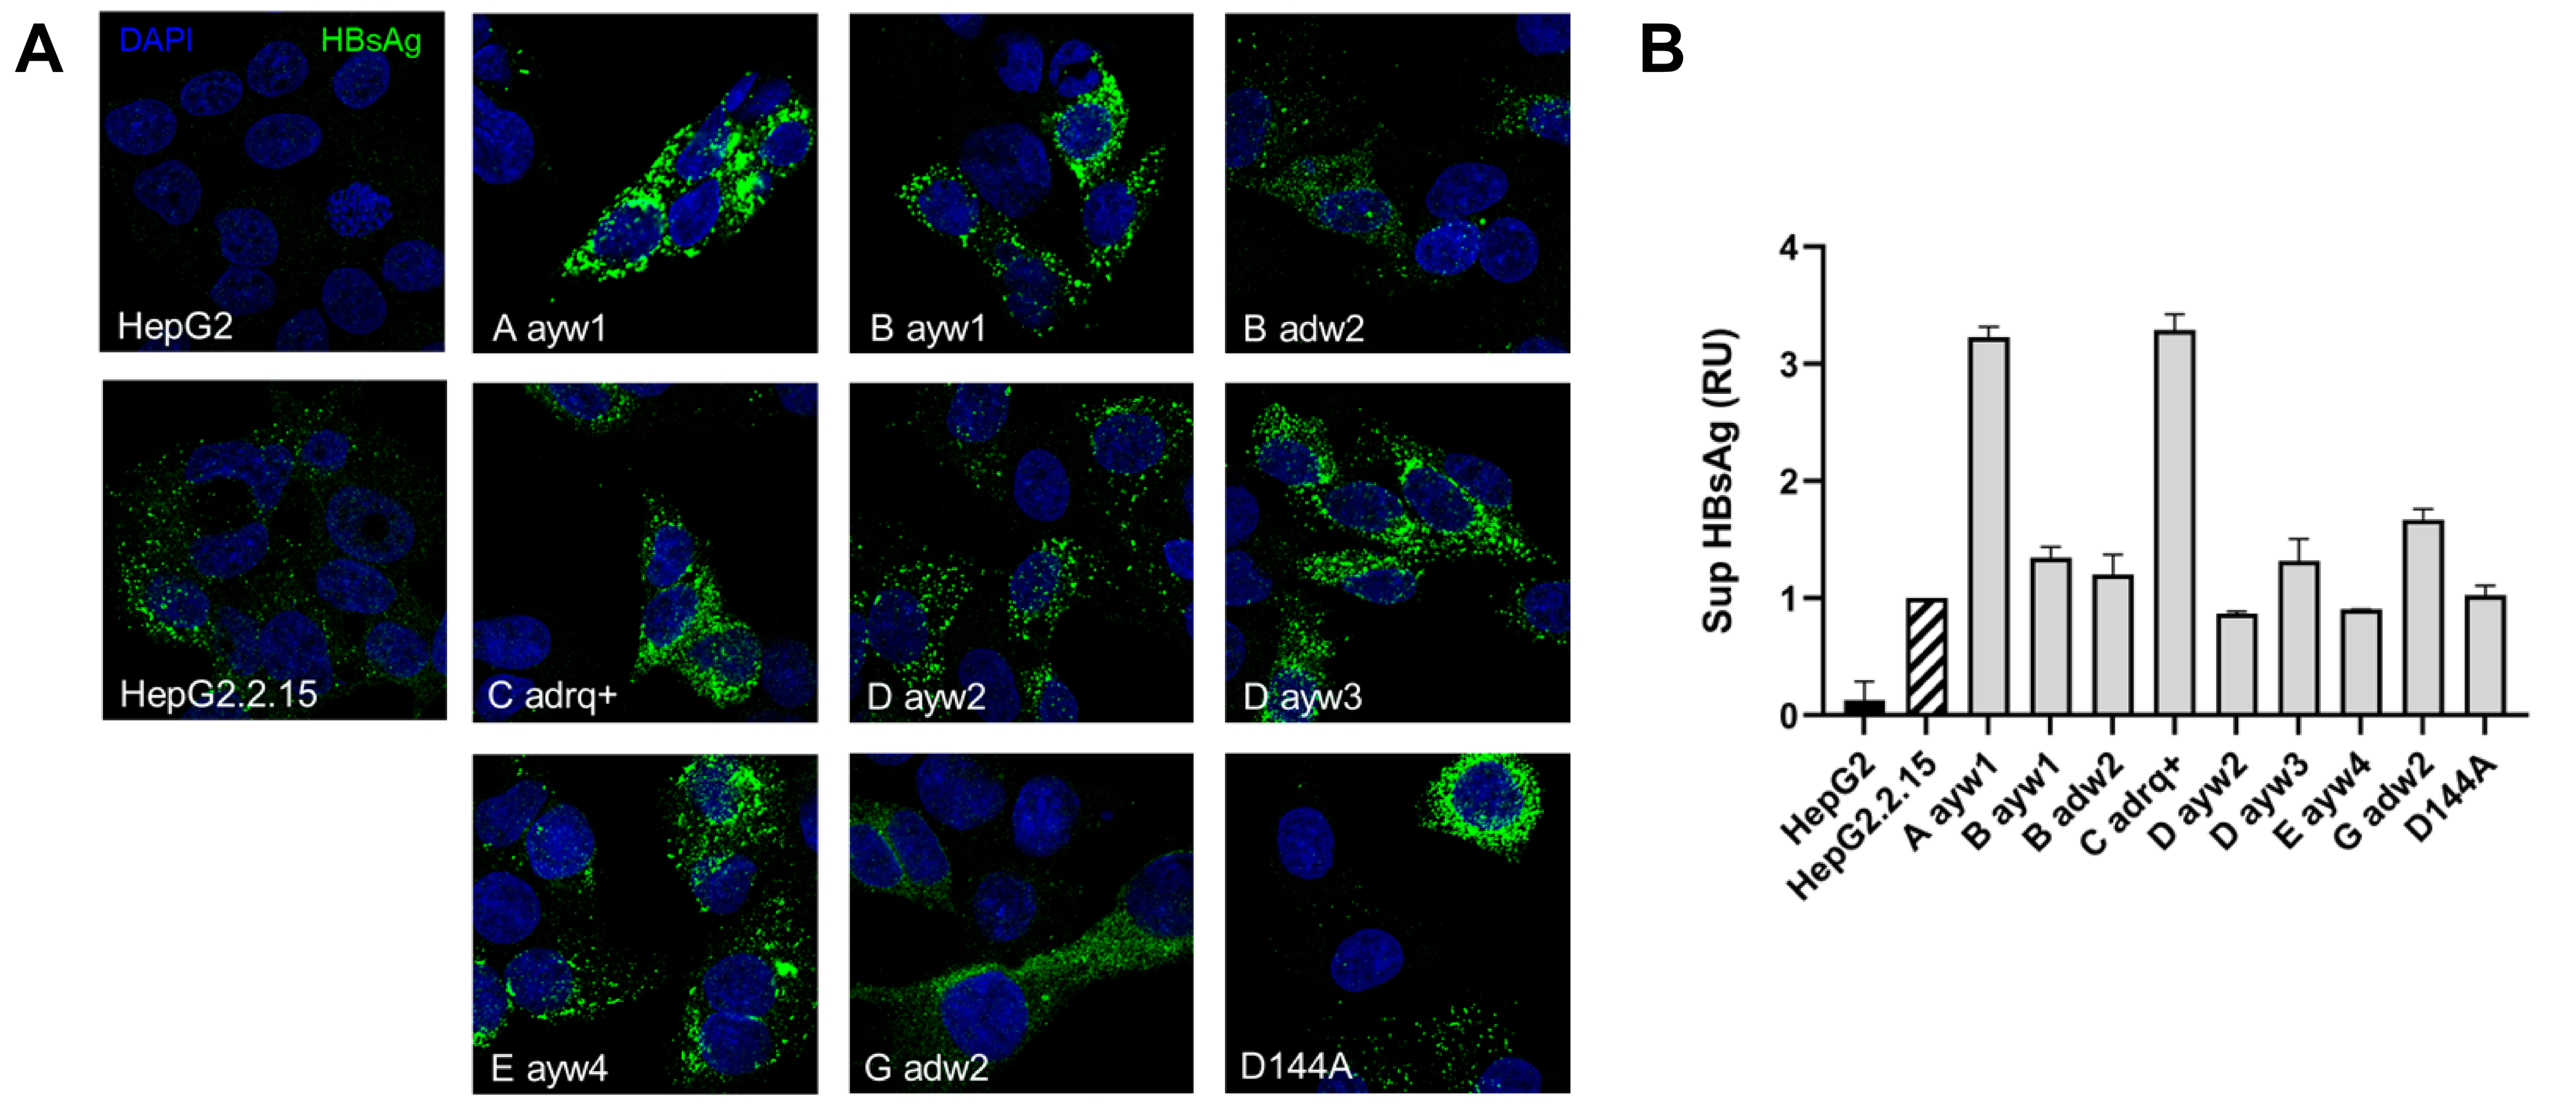

A
DAPI
HBsAg
HepG2
A ayw1
B ayw1
B adw2
HepG2.2.15
C adrq+
D ayw2
D ayw3
E ayw4
G adw2
D144A
B
Sup HBsAg (RU)
0
1
2
3
4
HepG2
HepG2.2.15
A ayw1
B ayw1
B adw2
C adrq+
D ayw2
D ayw3
E ayw4
G adw2
D144A

Fig 4. Antiviral effect of REP 2139 in GCLs and HepG2.2.15 cells.
(A) Experimental design is as indicated. (B) Chemical sequence of the NAP REP 2139. (C) Comparative antiviral effect of REP 2139 on HBsAg secretion for all indicated cell lines normalized to cell viability (BCA) and to 0 nM REP 2139. Sup, supernatant; RU, relative unit.

A

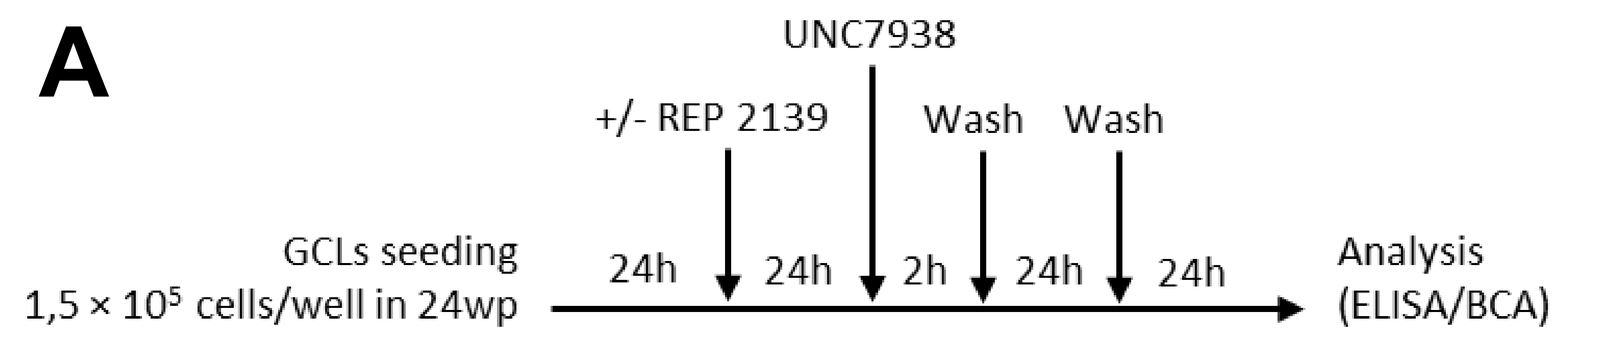

UNC7938
+/- REP 2139
Wash
Wash
GCLs seeding
1,5 × 10⁵ cells/well in 24wp
24h
24h
2h
24h
24h
Analysis
(ELISA/BCA)

B

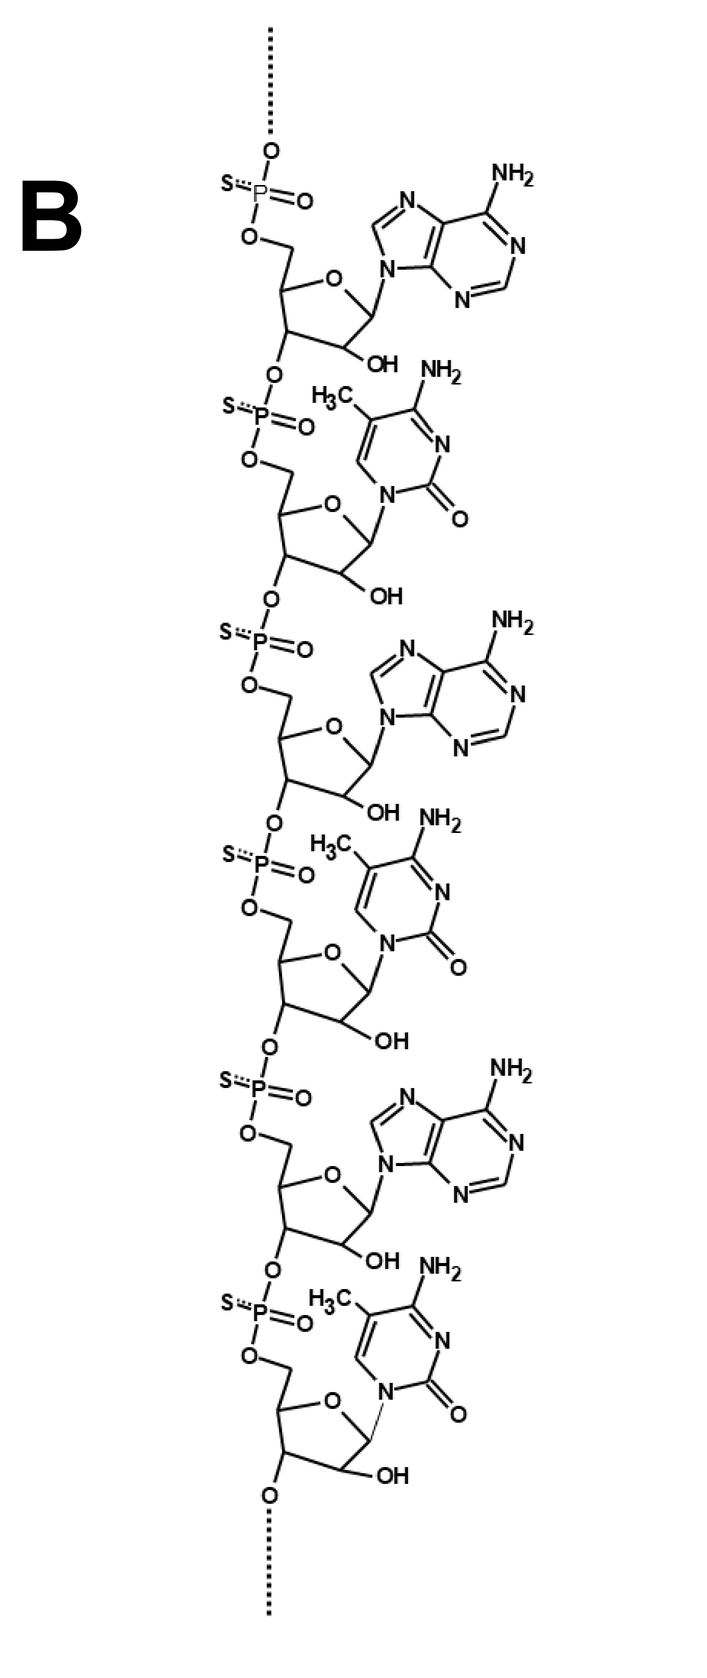

REP 2139
(2'OMeA,2'OMe-5-MeC)$_{20}$

C

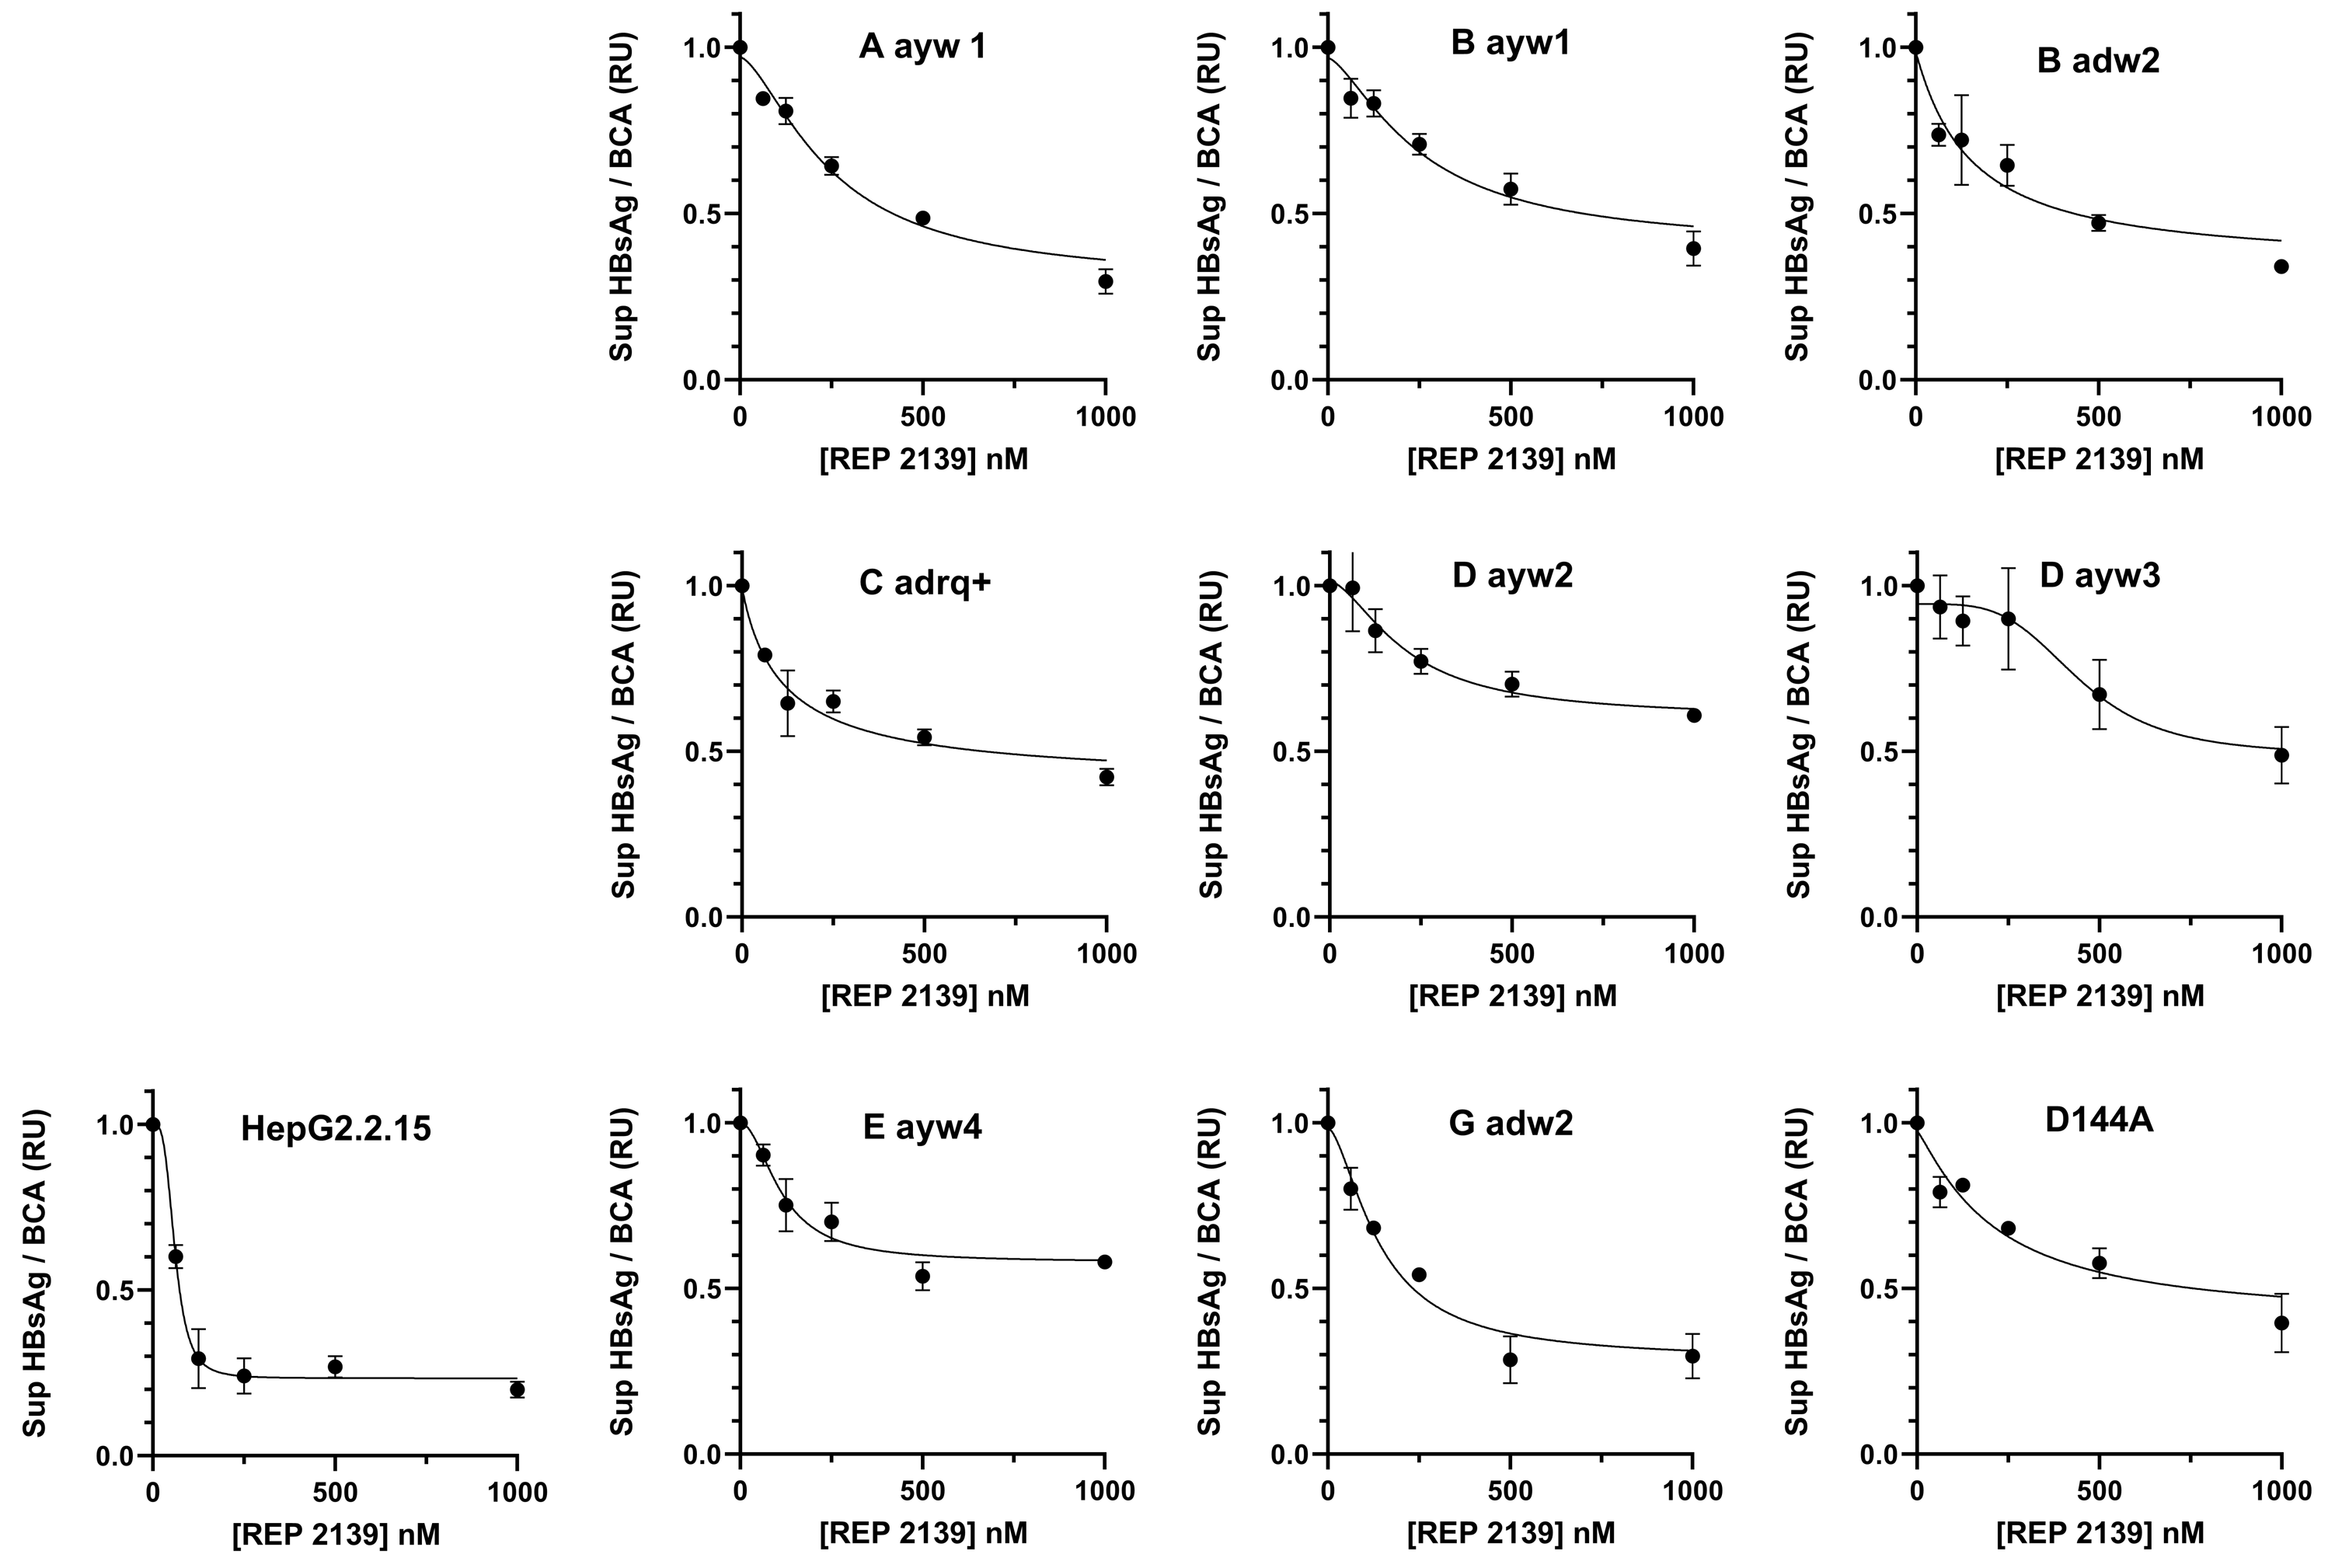

A ayw 1
B ayw1
B adw2
C adrq+
D ayw2
D ayw3
HepG2.2.15
E ayw4
G adw2
D144A
Sup HBsAg / BCA (RU)
[REP 2139] nM
0.0
0.5
1.0
0
500
1000

CONCLUSIONS

- ➢ **CRISPR/Cas9 was used to create in vitro HepG2-derivative cell lines expressing HBsAg from various genotypes and serotypes.**
- ➢ **REP 2139 has a pangenomic antiviral effect.**
- ➢ **REP 2139 has an antiviral effect in D144A vaccine escape mutant.**

REFERENCES

1. Blanchet et al., Antiviral Res 2019; 164: 97-105
2. Boulon et al., Antiviral Res 2020; 180: 104853
3. Roehl et al., Mol Ther Nuc Acids Res. 2017; 8: 1-12
4. Quinet et al., Hepatology 2018; 67: 2127-2140
5. Al-Mahtab et al., PLoS ONE 2016; 11: e0156667
6. Vaillant, ACS Inf Dis 2019: 5: 675-687

CONTACT

Dr. Patrick Labonté patrick.labonte@inrs.ca **Dr. Andrew Vaillant** availlant@replicor.com